BND-22, a first-in-class, anti–ILT2 monoclonal antibody inhibits the immunosuppressive effects of HLA-G and enhances anti-tumor activity of immune cells in preclinical in vitro, ex vivo, and in vivo models

Authors: Ilana Mandel, Dror Alishekevitz, Itay Friedman, Sharon Hashmueli, Dana Haves, Ilana Oz, Tsuri Peretz, Yair Sapir, Tehila Ben Moshe. **Affiliation:** Biond Biologics , Misgav, Israel

BIOND BIOLOGICS
Visit our website
Poster # 3266
AACR 2020

1 Background

ILT2 is a multi-cell inhibitory immune checkpoint which is expressed on innate and adaptive immune cells. It has been shown to bind to MHC class I molecules and binds with the highest affinity to HLA-G, an immunosuppressive MHC molecule expressed by multiple tumor types. ILT2-mediated inhibition can lead to impairment of cytotoxicity and proliferation of T cells and NK cell and generates a "don't eat me" signal, which prevents the efficient engulfment of tumor cells by macrophages identifying the ILT2 signaling axis as a potential novel target for anti-cancer immunotherapy.

BND-22 is a humanized IgG4 anti-ILT2 blocking antibody with high affinity and selectivity to ILT2. It was demonstrated that BND-22 can block ILT2-HLA-G interactions. ILT2 blockade with BND-22 increased macrophage-mediated phagocytosis of tumor cell lines and patient-excised tumors and enhanced the activity of T cells and NK cells against multiple types of cancer cells. ILT2 and PD-1 were found to be expressed by different sub-populations of CD8 T cells. Co-blockade of ILT2 and PD-1 showed a synergistic effect in increasing pro-inflammatory cytokine production by T cells. Various in vivo tumor xenograft models demonstrated the clear anti-tumor activity of **BND-22** which is mediated by its ability to increase the immune activation of both myeloid cells and lymphocytes.

Collectively, pre-clinical data demonstrated that **BND-22**, a first-in-class, anti-ILT2 antagonist antibody, can induce a potent macrophage and lymphocyte-driven anti-tumor immune response in multiple in vitro, ex vivo and in vivo models. **BND-22** offers a differentiated, multi immune cell-based mechanism of action. Safety, tolerability, and anti-tumor activity of **BND-22** will be explored in a first-in-human clinical trial in cancer patients with tumor types known to express HLA-G.

2 BND-22 activates both innate and adaptive immune cells, leading to a broad anti-tumor immune response

BND-22 enhances the ability of macrophages to phagocytose tumor cells

BND-22 enhances anti-tumor activity of T cells and NK cells

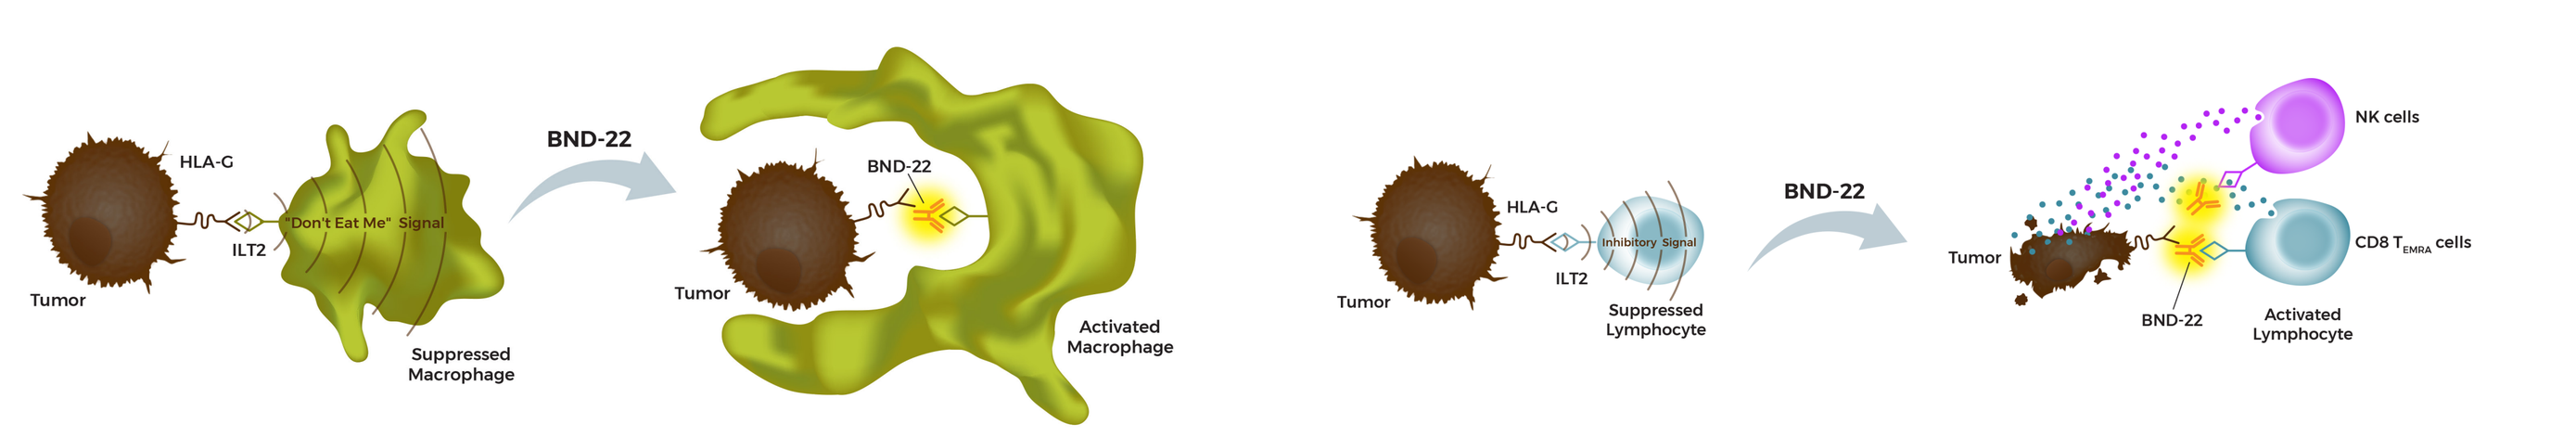
HLA-G
ILT2
Tumor
"Don't Eat Me" Signal
Suppressed Macrophage
BND-22
Activated Macrophage
Inhibitory Signal
Suppressed Lymphocyte
NK cells
CD8 TEMRA cells
Activated Lymphocyte

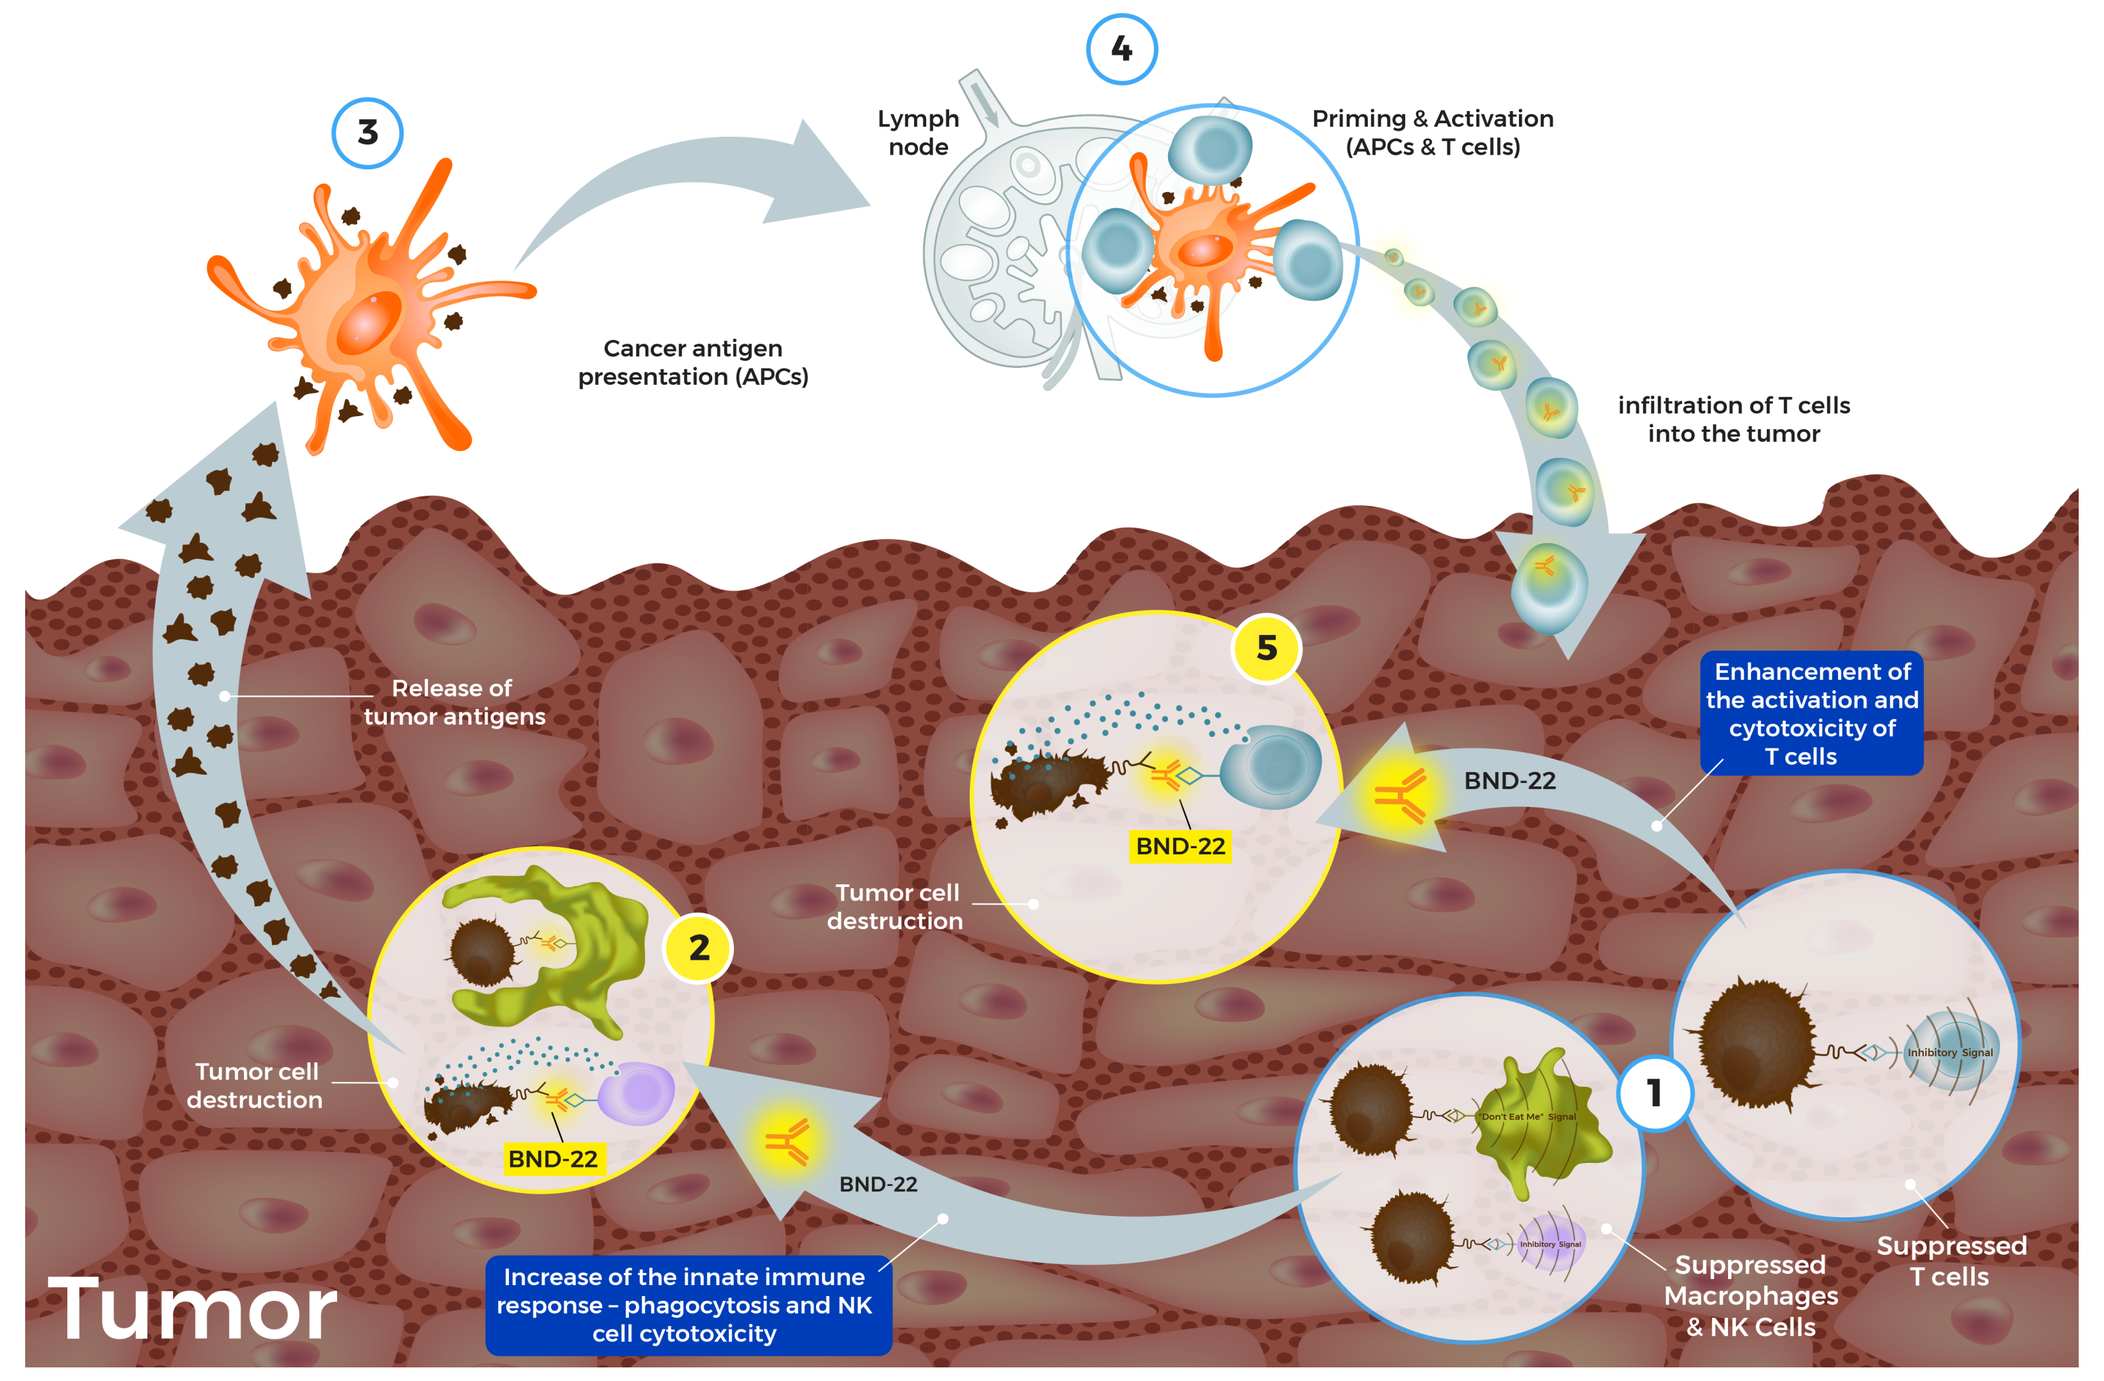
Lymph node
Priming & Activation (APCs & T cells)
Cancer antigen presentation (APCs)
infiltration of T cells into the tumor
Release of tumor antigens
Enhancement of the activation and cytotoxicity of T cells
BND-22
Tumor cell destruction
Increase of the innate immune response – phagocytosis and NK cell cytotoxicity
Suppressed Macrophages & NK Cells
Suppressed T cells
Tumor

Illustration of the MOA of BND-22: The tumor microenvironment contains macrophages, NK cells and T cells that are inhibited through HLA-G-ILT2 interactions (1). Once BND-22 is added, the antibody increases the innate immune response by enhancing both phagocytosis by macrophages and cytotoxicity of NK cells leading to tumor cell destruction and release of tumor antigens (2). These tumor antigens are released and are presented by antigen presenting cells in the periphery (3) which leads to T cell priming, activation and eventually infiltration into the tumor (4). In parallel, BND-22 acts directly on T cells to increase their activation and cytotoxicity in the tumor microenvironment (5). Taken together, this demonstrates the ability of BND-22 to generate a broad anti-tumor response which is mediated by its ability to activate innate and adaptive immune cells

3 BND-22 binds ILT2 and blocks ILT2-HLA-G interactions

A. BND-22 binds rILT2 with high affinity

Ka (1/Ms)	Kd (1/s)	KD (/M)
1.37-1.76 E+06	2.22-5.22 E-03	1.26-3.16 E-09

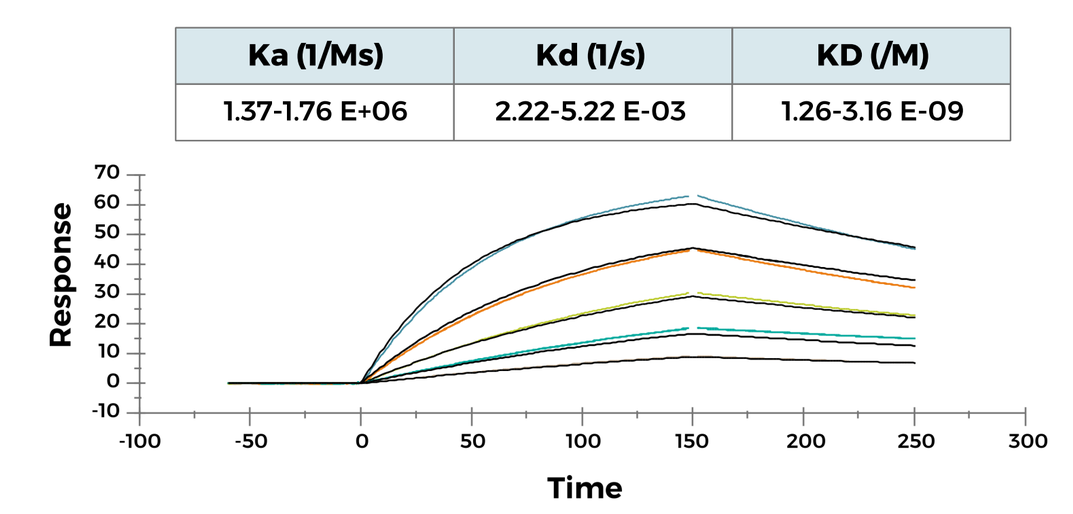
Response
Time

B. BND-22 binds membranal ILT2

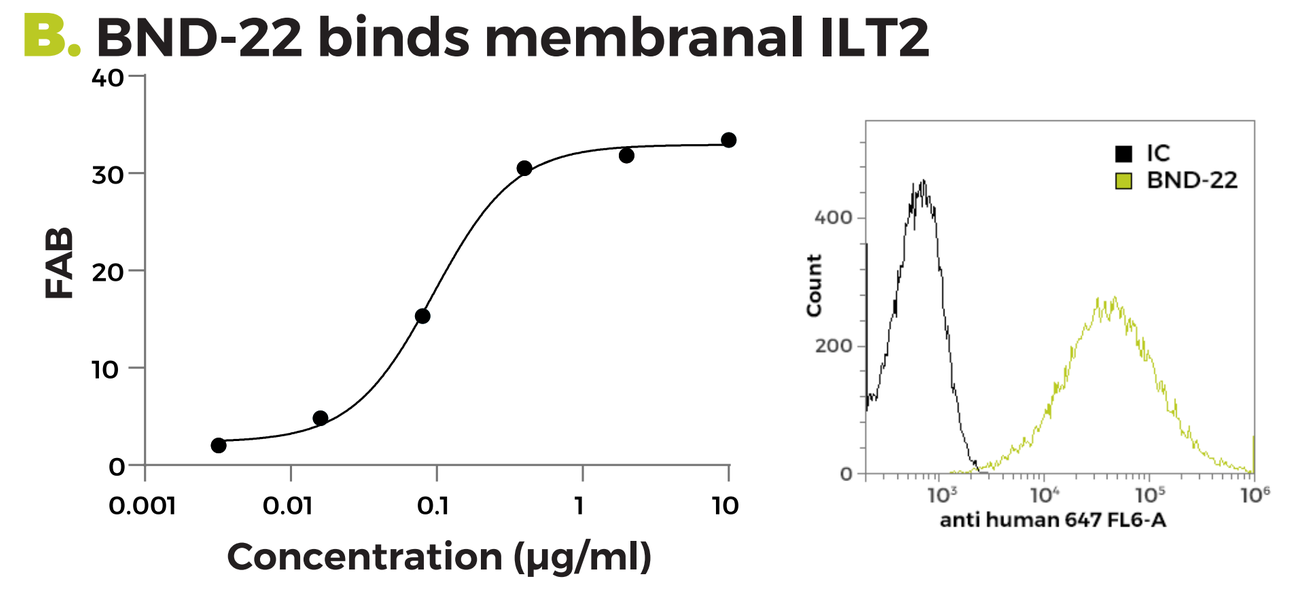
FAB
Concentration (µg/ml)
IC
BND-22
Count
anti human 647 FL6-A

C. BND-22 blocks binding of rILT2 to HLA-G+ cells - FACS

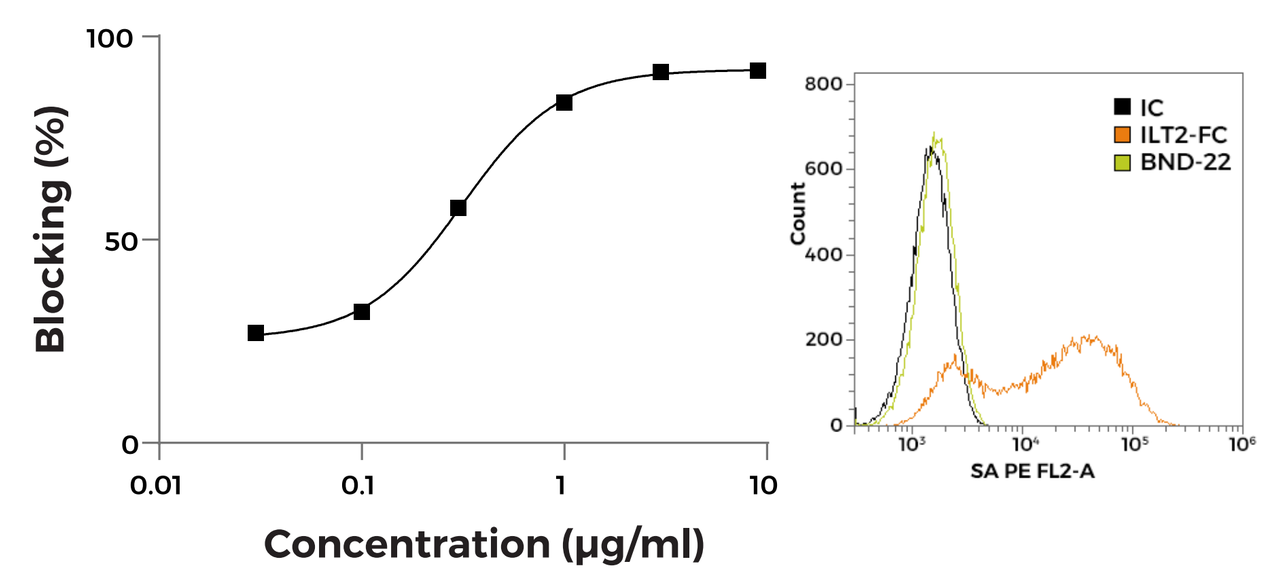
Blocking (%)
Concentration (µg/ml)
IC
ILT2-FC
BND-22
Count
SA PE FL2-A

D. BND-22 blocks binding of rILT2 to HLA-G+ cells – reporter assay

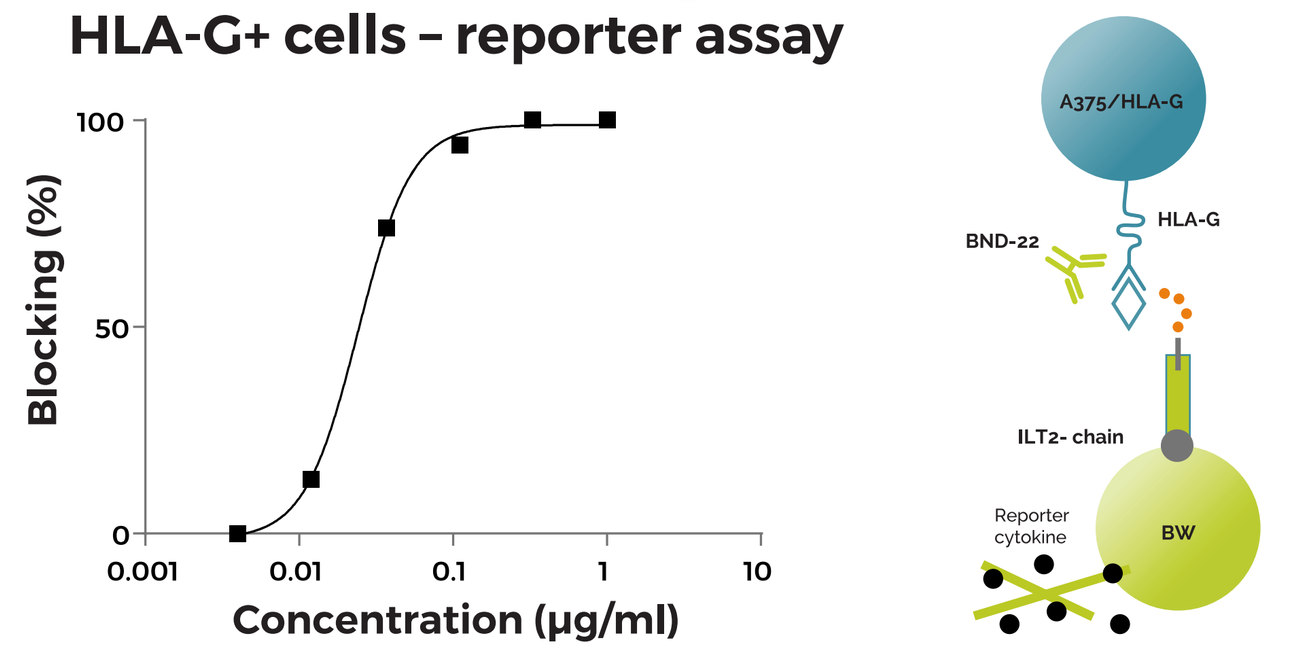
Blocking (%)
Concentration (µg/ml)
A375/HLA-G
HLA-G
BND-22
ILT2- chain
Reporter cytokine
BW

A. Biacore analysis of BND-22 binding to recombinant ILT2-HIS. B. FACS analysis of binding to ILT2 transfected BW cells. C. Blocking FACS with recombinant ILT2 and HLA-G+ cells in the presence of BND-22. D. Cell- based reporter assay to detect BND-22 mediated blocking of HLA-G-ILT2 interactions as determined by secretion of a reporter cytokine.

4 ILT2 is highly expressed in the TME and periphery of cancer patients

ILT2 is highly expressed in peripheral lymphocytes of cancer patients

A.

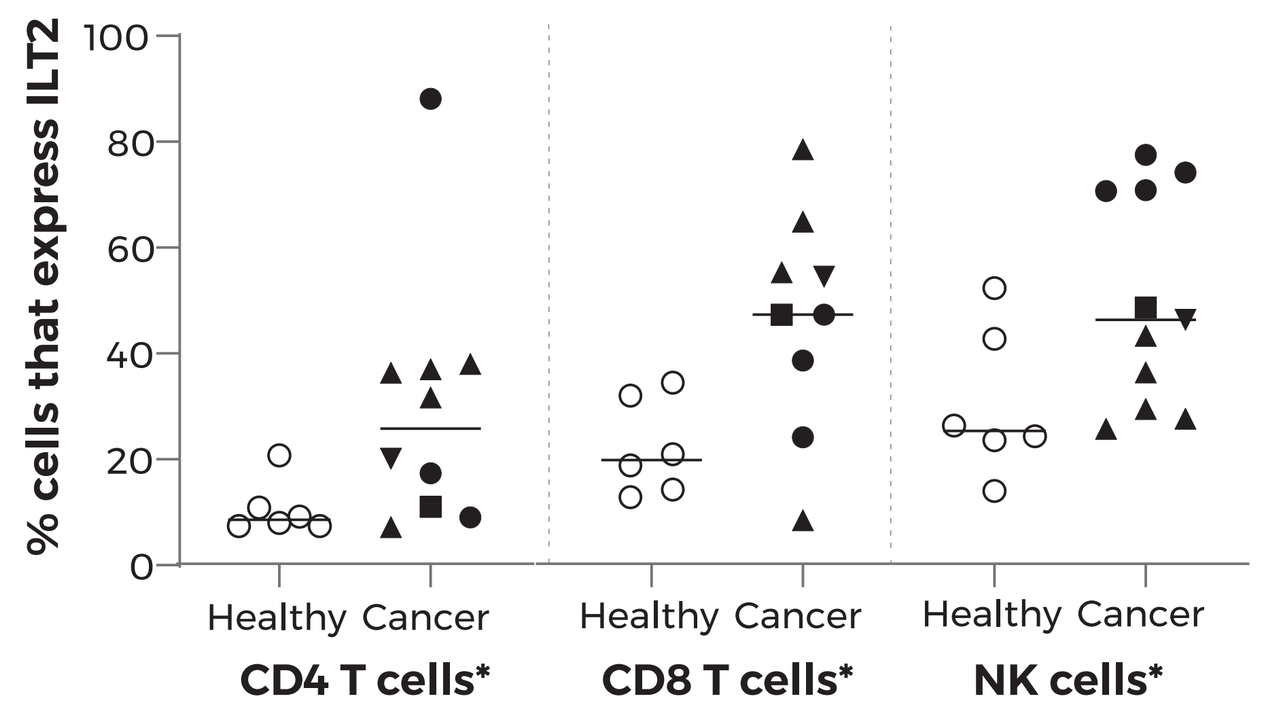
% cells that express ILT2
Healthy Cancer
CD4 T cells*
CD8 T cells*
NK cells*

ILT2 is expressed in the TME by innate and adaptive immune cells

B.

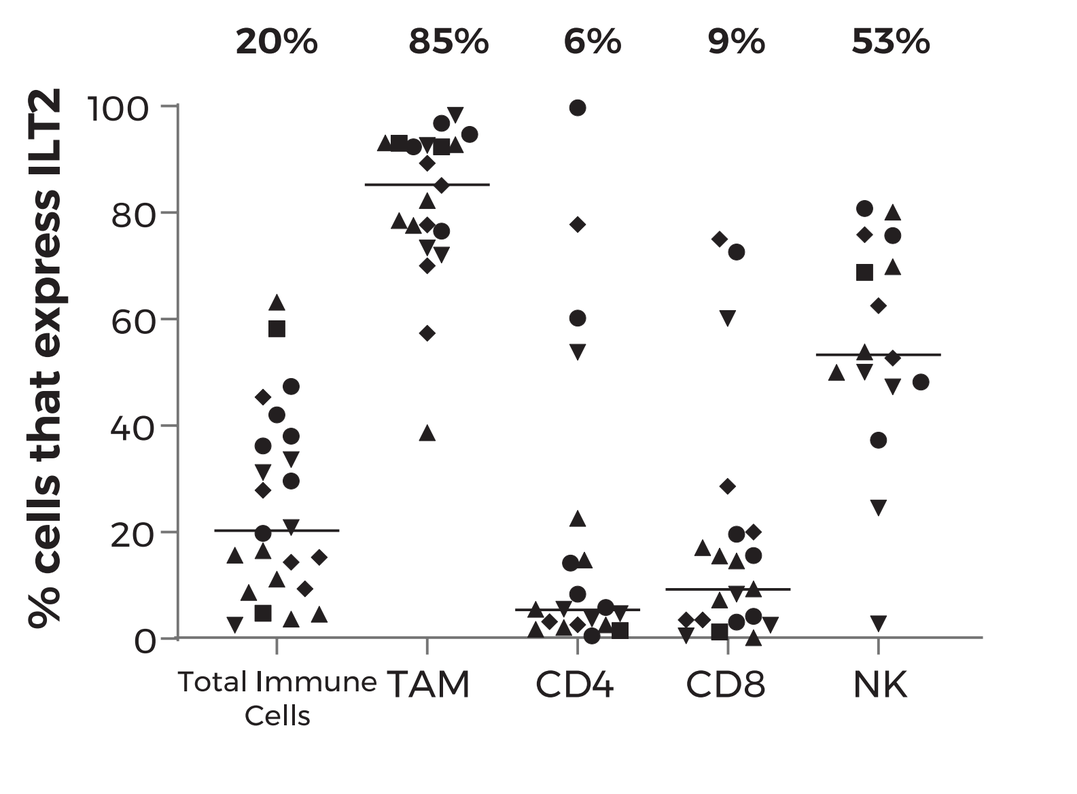
20%
85%
6%
9%
53%
% cells that express ILT2
Total Immune Cells
TAM
CD4
CD8
NK

C.

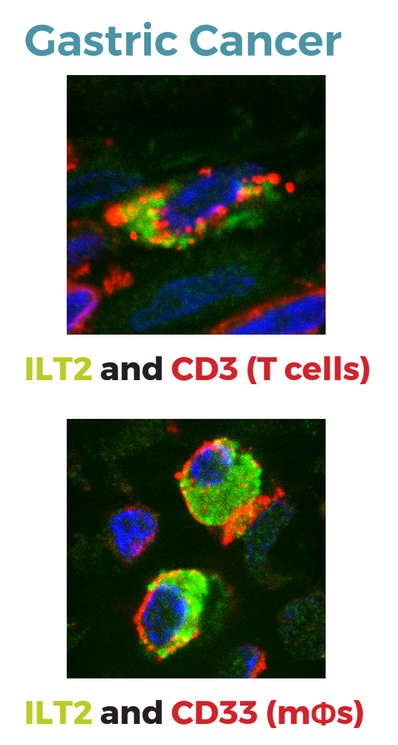
Gastric Cancer
ILT2 and CD3 (T cells)
ILT2 and CD33 (mΦs)

■ NSCLC ● Kidney (RCC) ▲ H&N ▼ Esophagus ◆ Colon ○ Healthy

A. PBMC were isolated from the blood of the indicated cancer patients or from healthy donors. The percent of ILT2+ NK cells, CD8 and CD4 T cells was evaluated by flow cytometry. *P<0.05; Student's T-test. B. The percent of ILT2+ in total intra-tumoral immune cells, Tumor associated macrophages (TAM), CD8, CD4 T cells and NK cells was evaluated by flow cytometry in fresh tumors isolated from operations of the indicated patients . The median of ILT2-positive cells for each immune cell population is indicated. C. ILT2 co-expression with T cells and myeloid cell markers in gastric cancer tumors as determined by immunofluorescence. TME - tumor microenviorment.

5 HLA-G is highly expressed in the TME and periphery of cancer patients

A. HLA-G is highly expressed in the TME of different cancer types and not in normal tissues

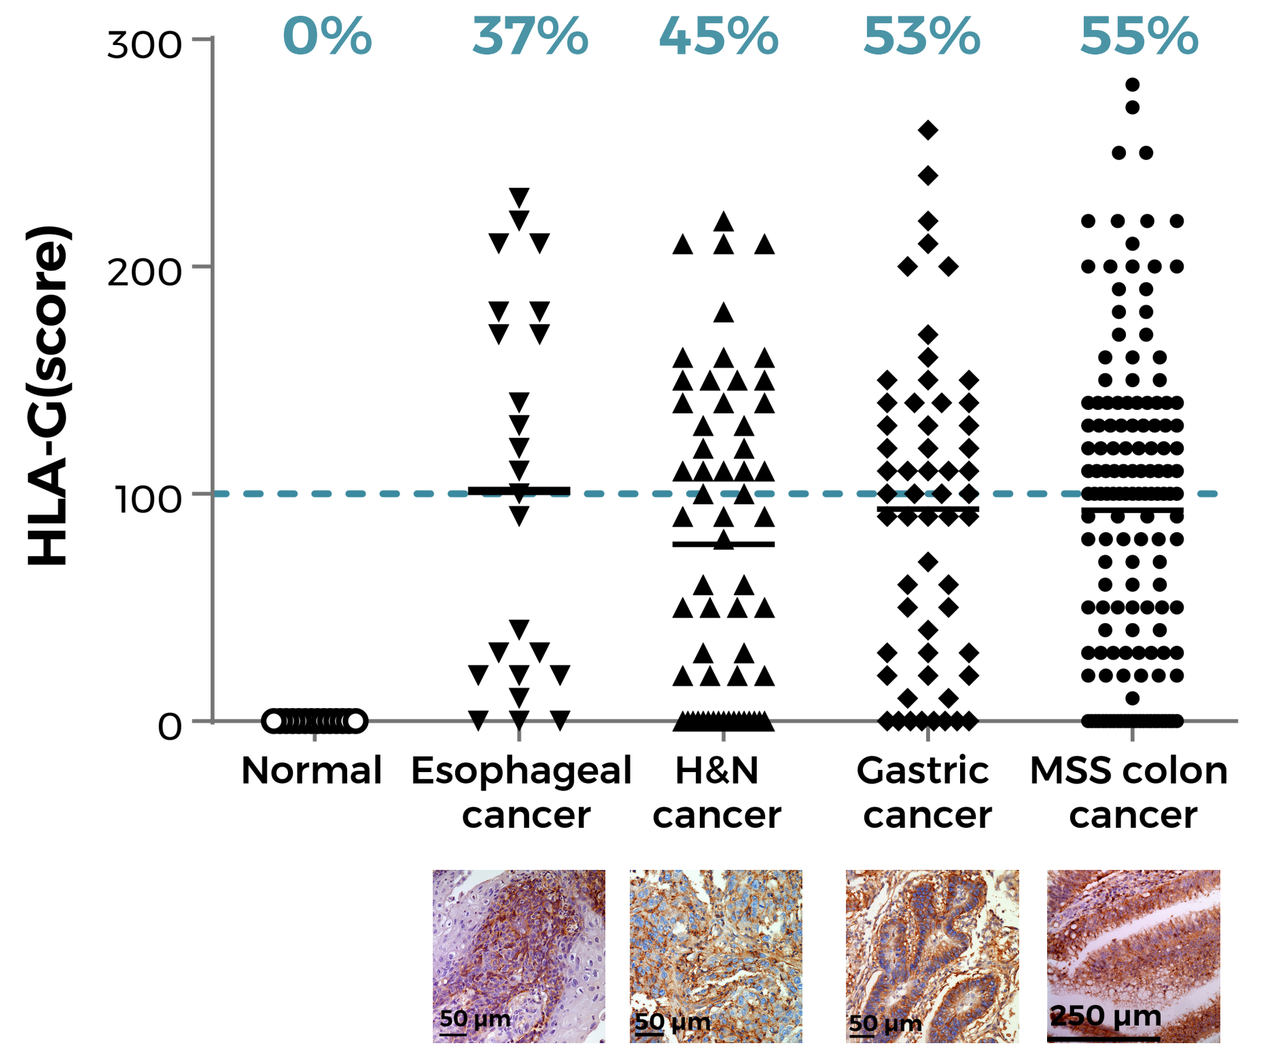
0%
37%
45%
53%
55%
HLA-G(score)
Normal
Esophageal cancer
H&N cancer
Gastric cancer
MSS colon cancer
50 µm
250 µm

B. Soluble HLA-G is highly expressed in the periphery of different cancer types

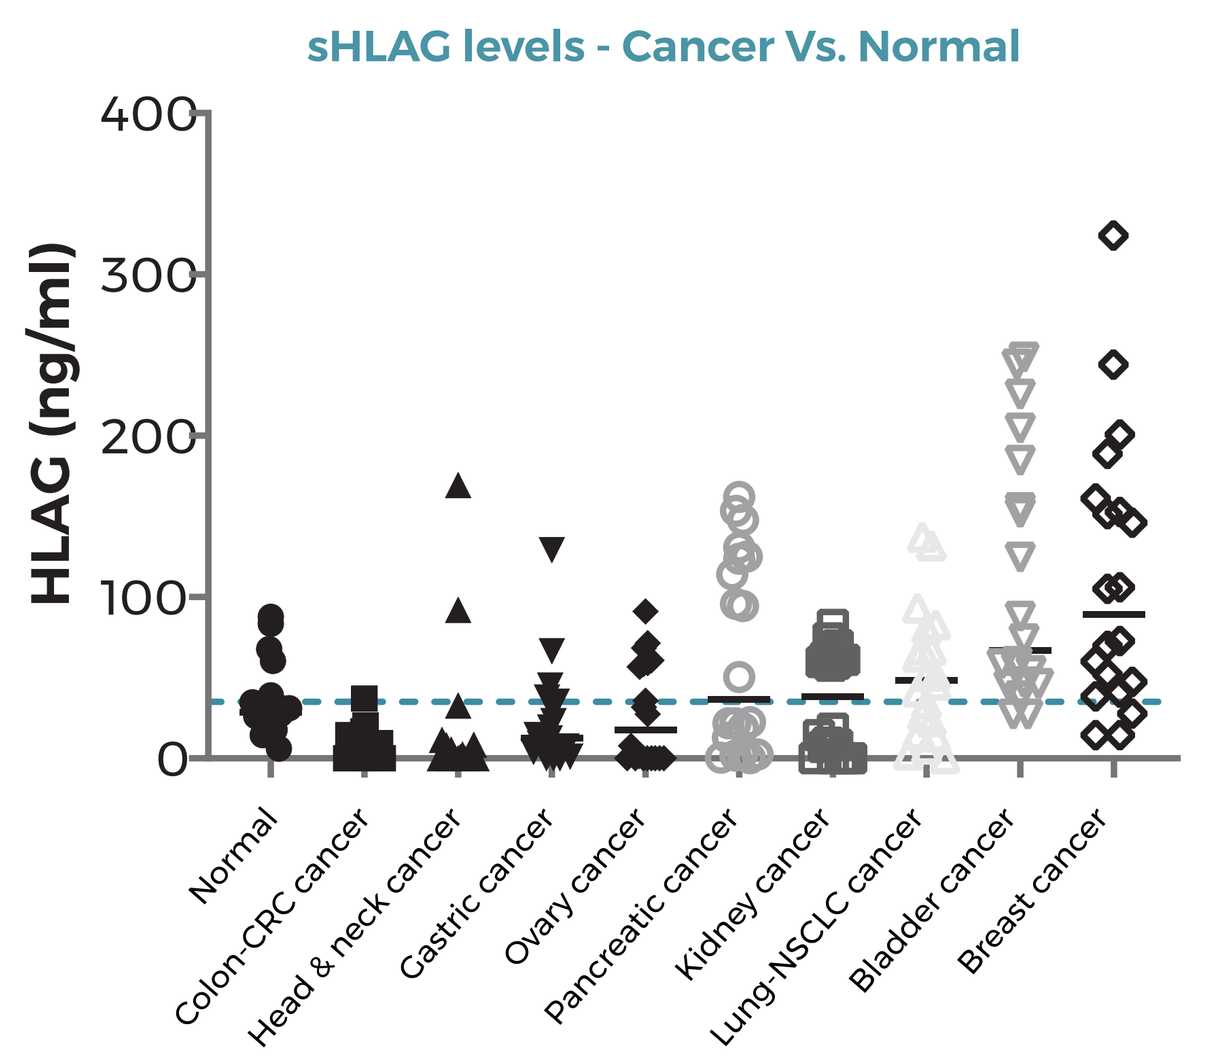
sHLAG levels - Cancer Vs. Normal
HLAG (ng/ml)
Normal
Colon-CRC cancer
Head & neck cancer
Gastric cancer
Ovary cancer
Pancreatic cancer
Kidney cancer
Lung-NSCLC cancer
Bladder cancer
Breast cancer

A. Tissue microarrays from the indicated cancer types were stained with anti-HLA-G. The percentages presented indicate the percent of cases which had a high staining score of above 100.
B. Soluble HLA-G was evaluated by ELISA in plasma samples from the indicated samples. Normal - H&N and esophageal tissues from healthy donors.

6 BND-22 enhances macrophages' phagocytosis of cancer cells

A. In vitro

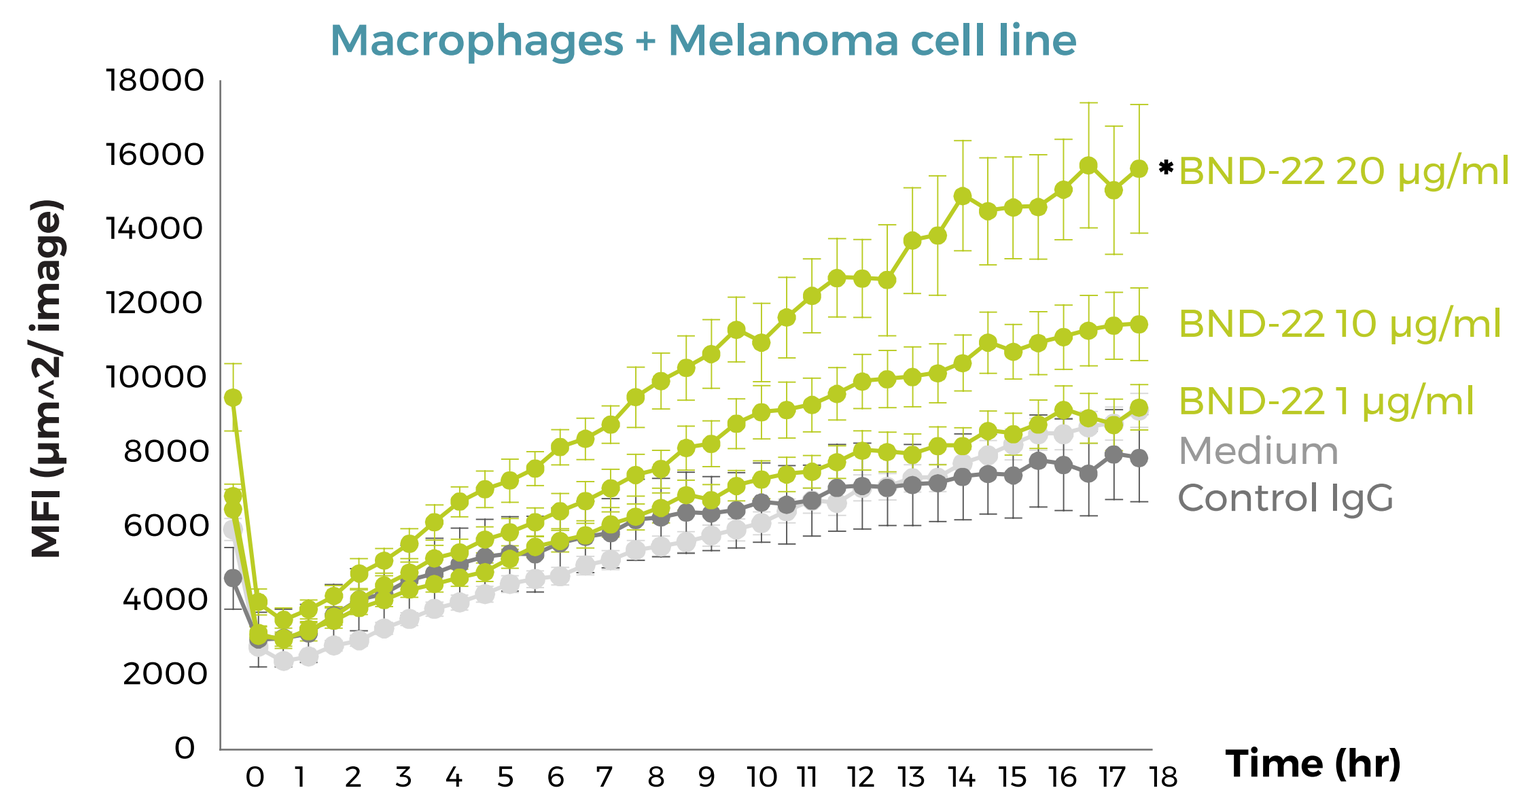
Macrophages + Melanoma cell line
MFI (µm^2/ image)
Time (hr)
BND-22 20 µg/ml
BND-22 10 µg/ml
BND-22 1 µg/ml
Medium
Control IgG

C. Ex vivo

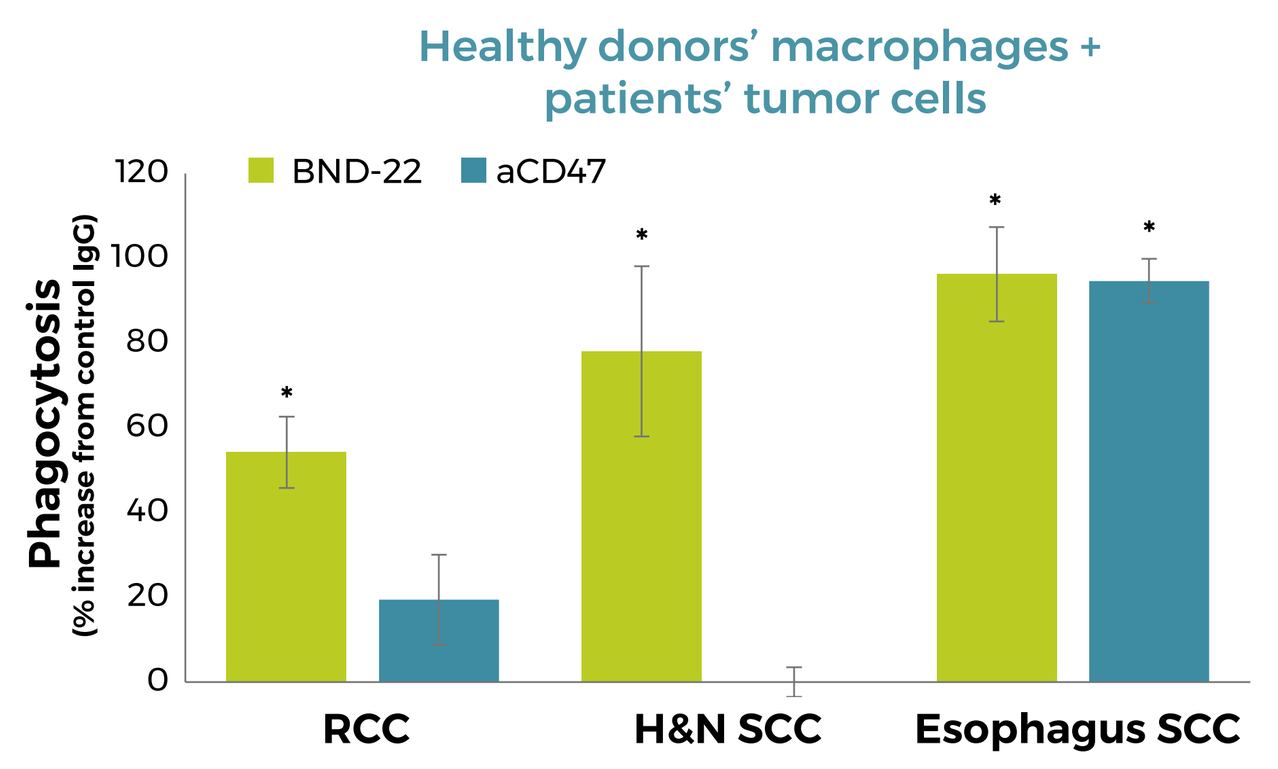
Healthy donors' macrophages + patients' tumor cells
BND-22
aCD47
Phagocytosis (% increase from control IgG)
RCC
H&N SCC
Esophagus SCC

B. Ex vivo

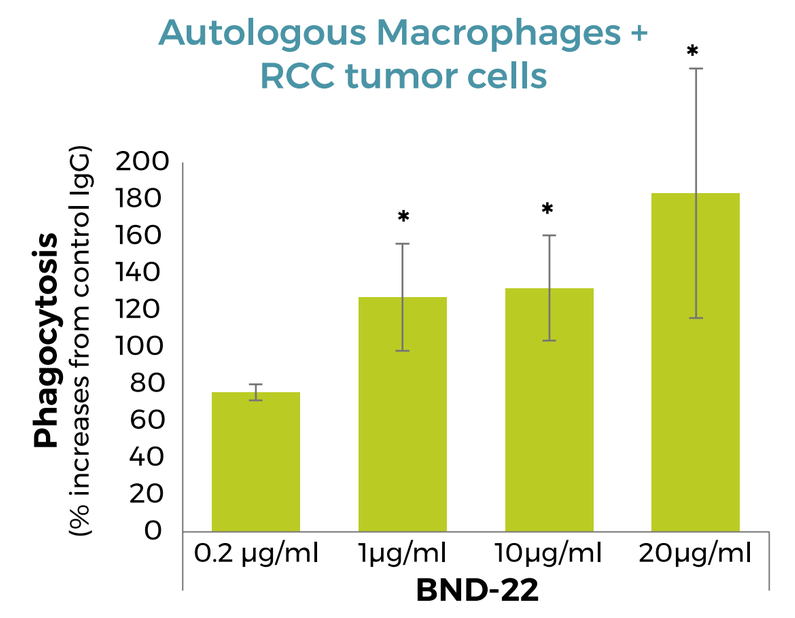
Autologous Macrophages + RCC tumor cells
Phagocytosis (% increase from control IgG)
0.2 µg/ml
1µg/ml
10µg/ml
20µg/ml
BND-22

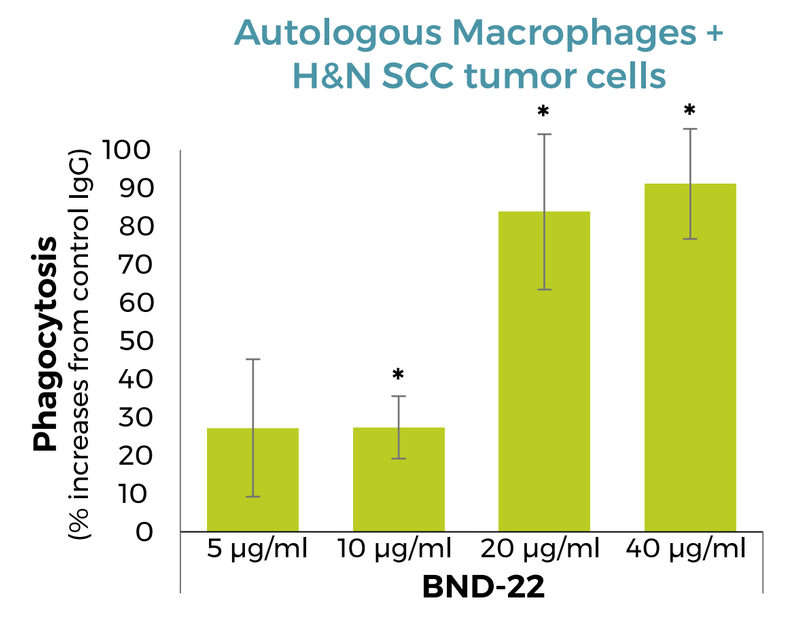
Autologous Macrophages + H&N SCC tumor cells
Phagocytosis (% increases from control IgG)
5 µg/ml
10 µg/ml
20 µg/ml
40 µg/ml
BND-22

A. Phagocytosis of A375 cells by macrophages generated from healthy donors. B,C. Phagocytosis of fresh tumor cells from the indicated patients that were added to macrophages generated from the same patients (B) or from healthy donors (C) . The percent of phagocytosis increase compared to control IgG at is presented (B+C). *P<0.05; Student's T-test compared to cells with control IgG.

7 BND-22 enhances T cell activity against cancer cells

ILT2 is highly expressed in a distinct CD8 T cell population – T_{EMRA} T cells

A.

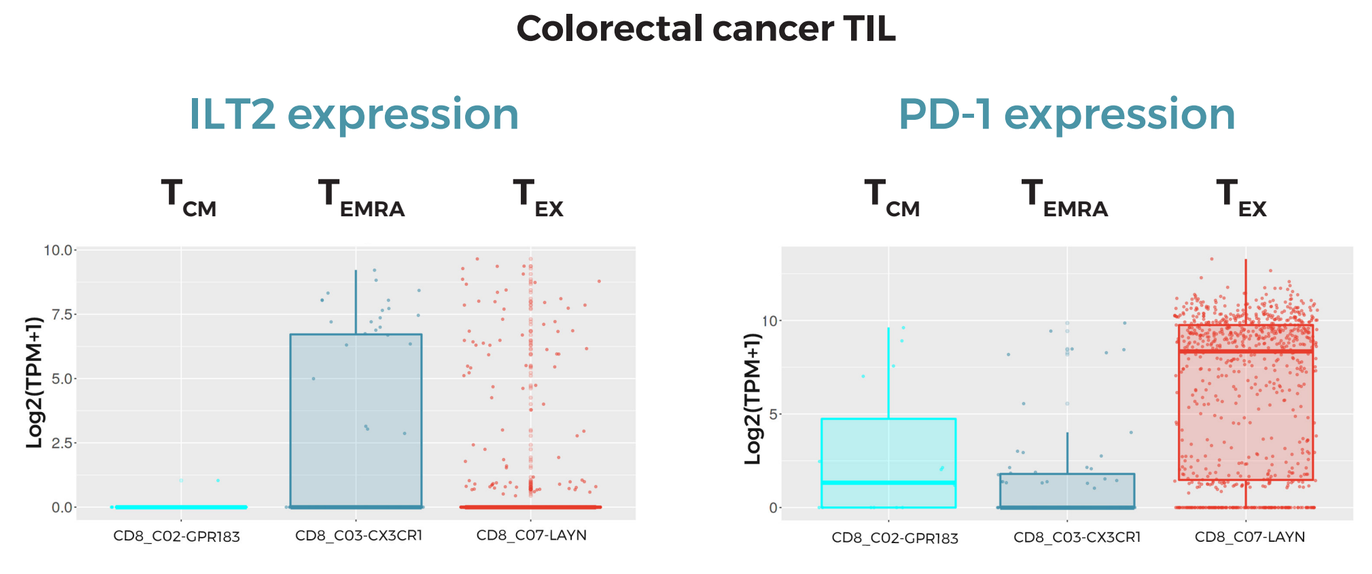
Colorectal cancer TIL
ILT2 expression
PD-1 expression
Log2(TPM+1)

B.

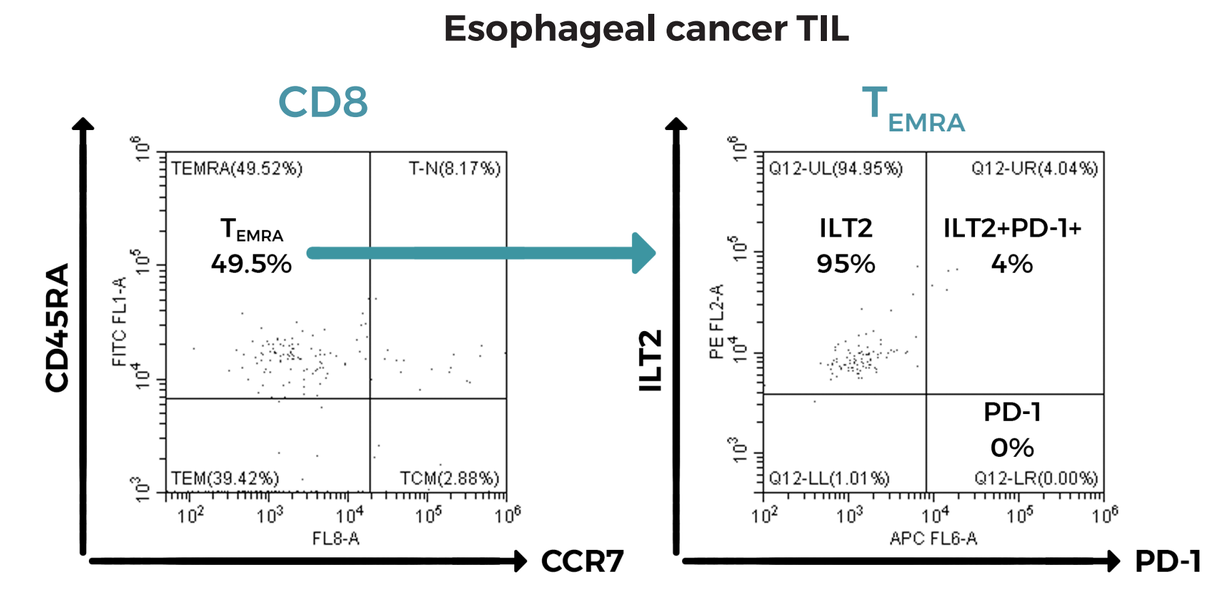
Esophageal cancer TIL
CD8
TEMRA 49.5%
CD45RA
CCR7
TEMRA
ILT2 95%
ILT2+PD-1+ 4%
PD-1 0%
ILT2
PD-1

Combining BND-22 with anti-PD-1 has a synergistic effect on T cell anti-tumor activity

C.

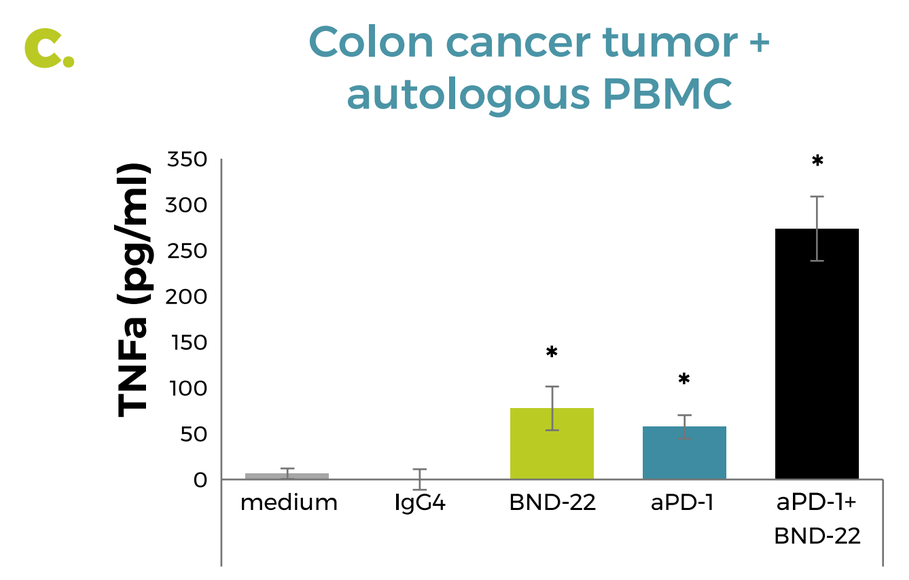
Colon cancer tumor + autologous PBMC
TNFa (pg/ml)
medium
IgG4
BND-22
aPD-1
aPD-1+ BND-22

D.

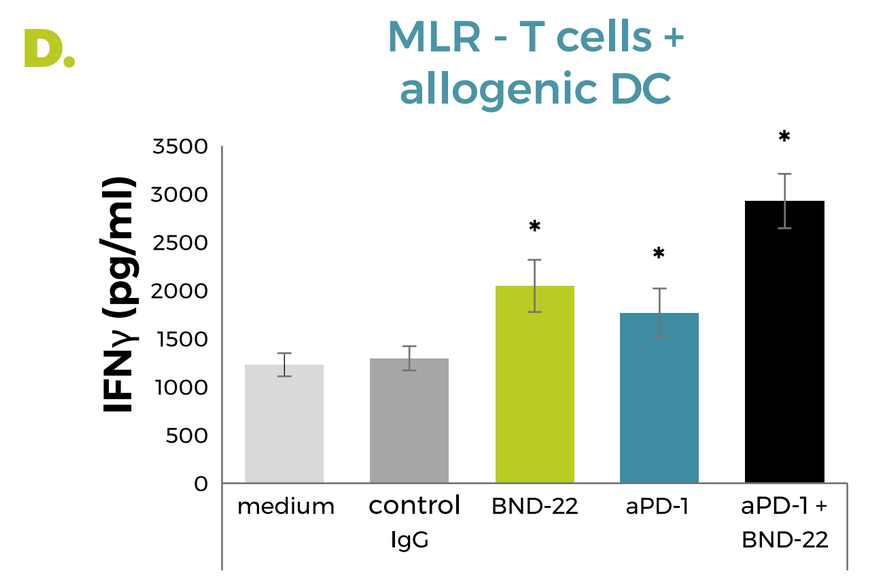
MLR - T cells + allogenic DC
IFNγ (pg/ml)
medium
control IgG
BND-22
aPD-1
aPD-1 + BND-22

E.

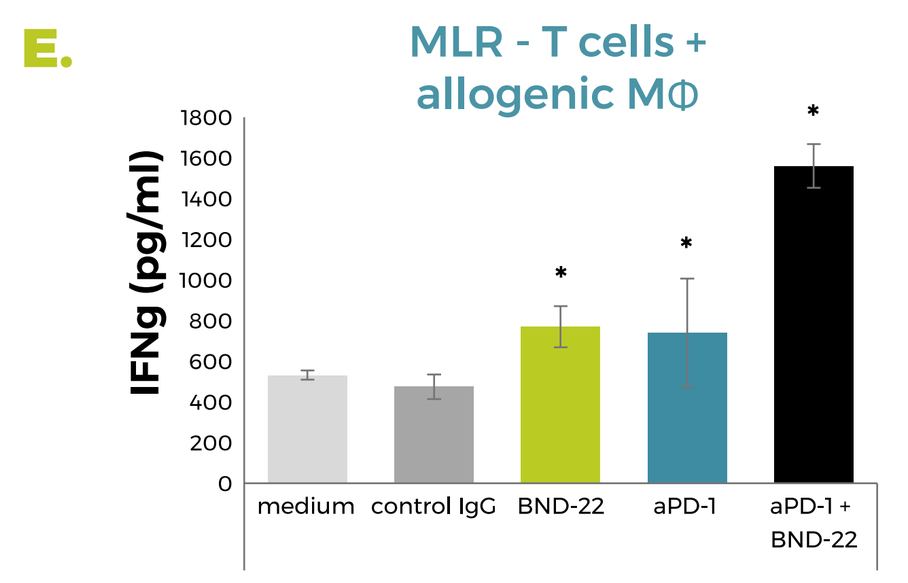
MLR - T cells + allogenic MΦ
IFNg (pg/ml)
medium
control IgG
BND-22
aPD-1
aPD-1 + BND-22

A. An analysis of ILT2 and PD-1 expression in intra-tumoral CD8 T cells from CRC patients from http://crctcell.cancer-pku.cn/. TCM – central memory T cells. TEMRA - T cells re-expressing CD45RA . TEX – exhausted T cells. B. Evaluation of ILT2 and PD-1 expression in intra-tumoral TEMRA CD8 T cells. C. Tumor cells from a colon cancer patient were incubated with PBMC generated from the same patient. D. Dendritic cells (DC) (D) or macrophages (E) were incubated with autologous CD8 T cells. TNFa (C) or IFNg secretion (D,E) were quantified by ELISA. *P<0.05; Student's T-test compared to control IgG.

8 BND-22 enhances NK cell activity against cancer cells

A.

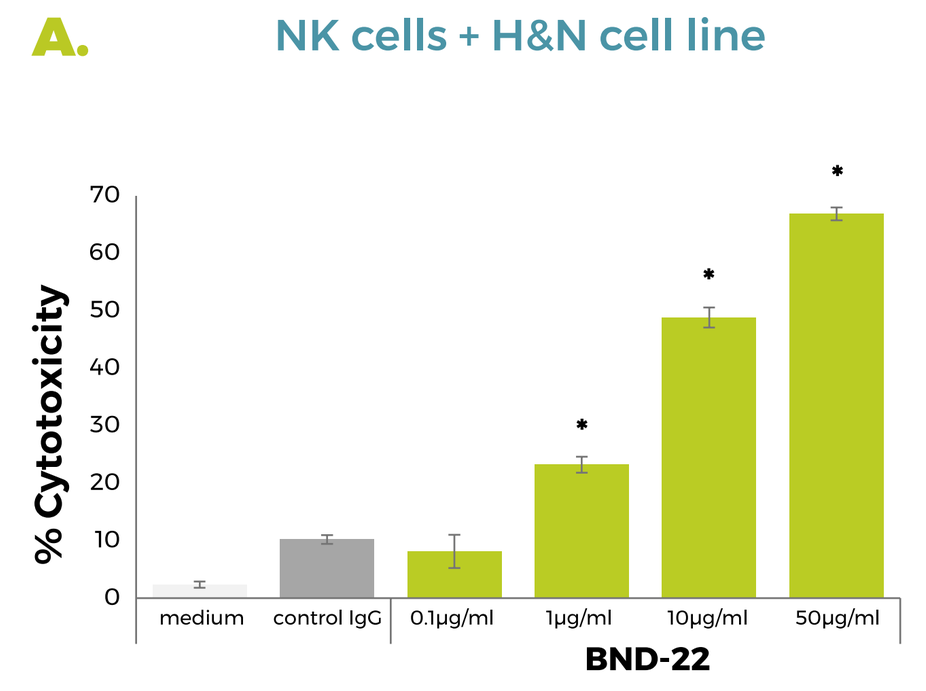
NK cells + H&N cell line
% Cytotoxicity
medium
control IgG
0.1µg/ml
1µg/ml
10µg/ml
50µg/ml
BND-22

B.

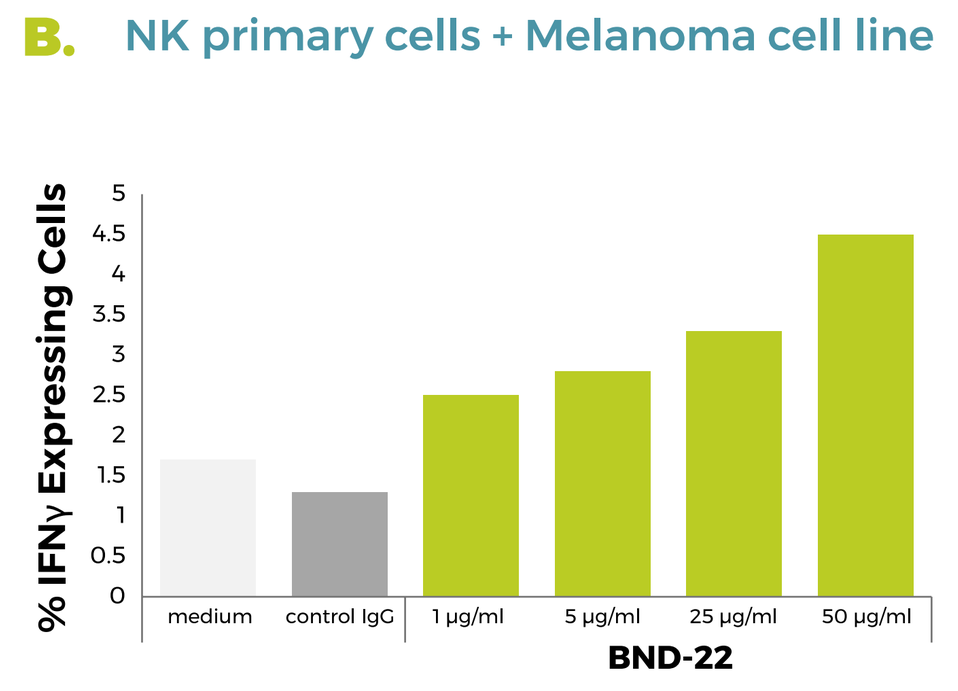
NK primary cells + Melanoma cell line
% IFNγ Expressing Cells
medium
control IgG
1 µg/ml
5 µg/ml
25 µg/ml
50 µg/ml
BND-22

C.

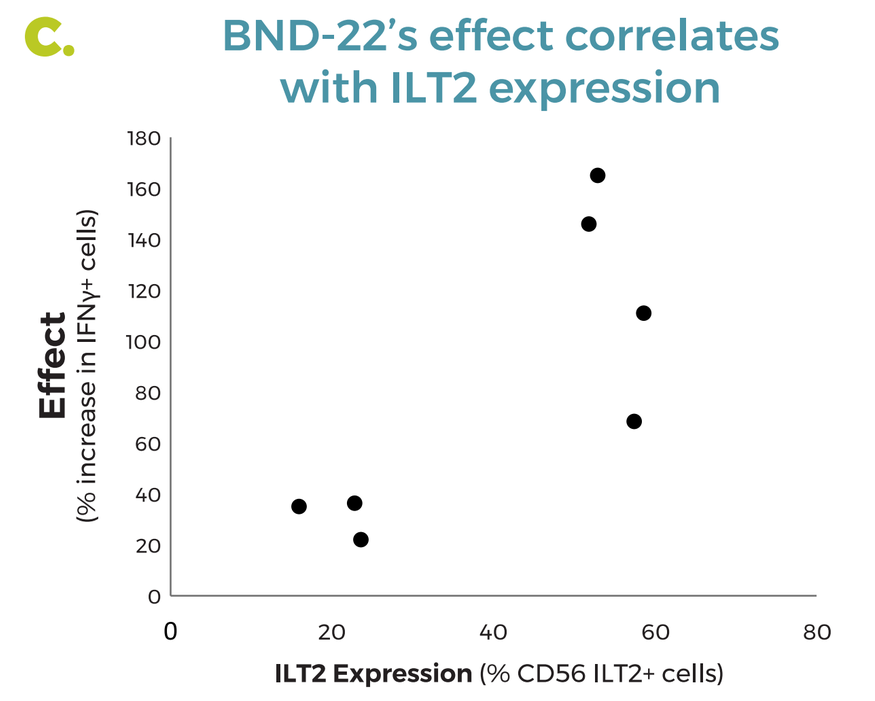
BND-22's effect correlates with ILT2 expression
Effect (% increase in IFNγ+ cells)
ILT2 Expression (% CD56 ILT2+ cells)

A. NK cell line cells were co-incubated with A253 cells followed by detection of cytotoxicity. B. NK cells isolated from healthy donors were co-incubated with A375 followed by flow cytometry analysis of intracellular IFNg. C. Correlation between ILT2 levels of NK cells and increase in IFNg presented in B.

9 BND-22 inhibits tumor growth *in vivo*

BND-22 enhances lymphocyte anti-tumor activity: in vivo melanoma lung lesion model

A. Prevention

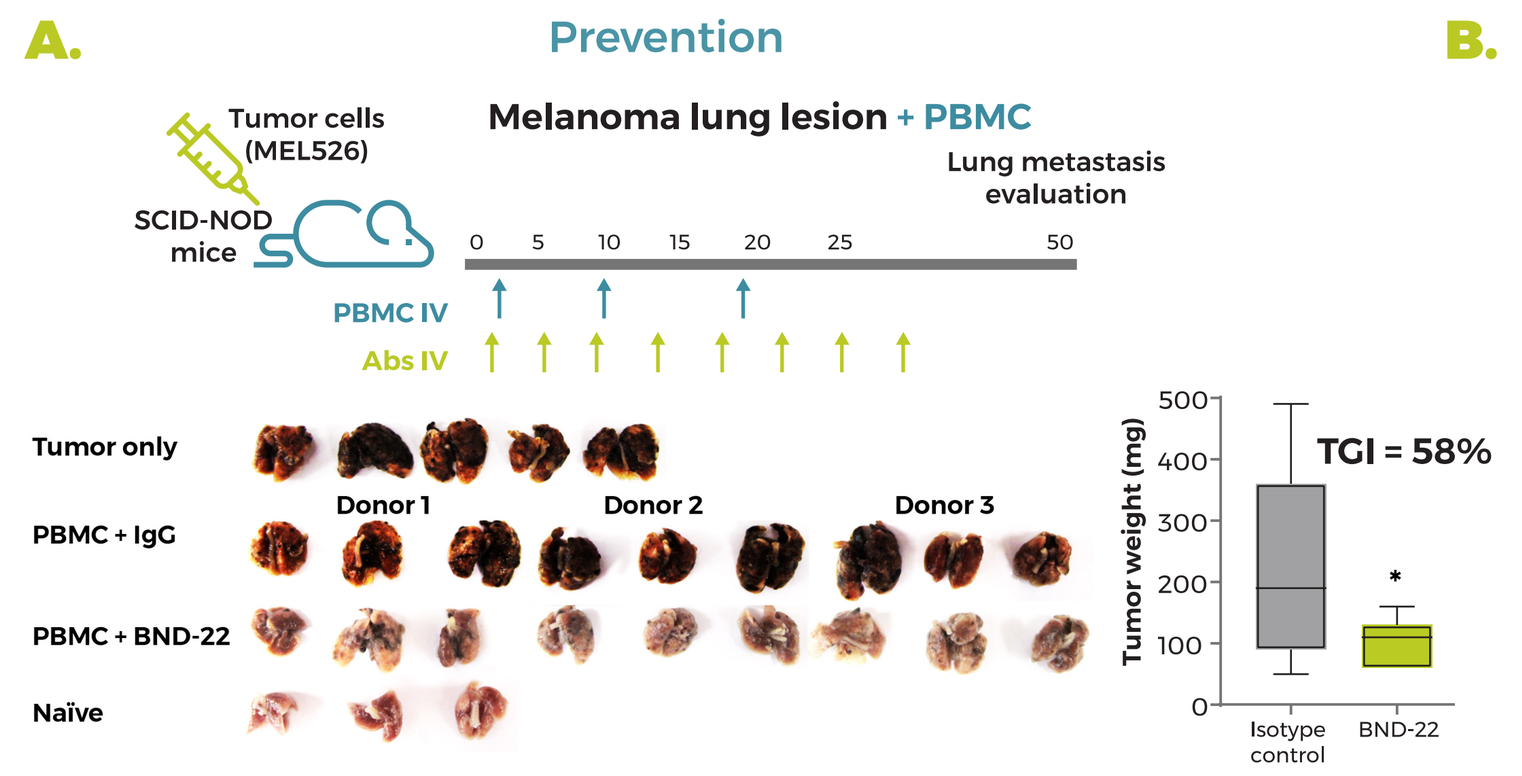
Tumor cells (MEL526)
SCID-NOD mice
Melanoma lung lesion + PBMC
Lung metastasis evaluation
PBMC IV
Abs IV
Tumor only
PBMC + IgG
PBMC + BND-22
Naïve
Donor 1
Donor 2
Donor 3
Tumor weight (mg)
TGI = 58%
Isotype control
BND-22

B. Established

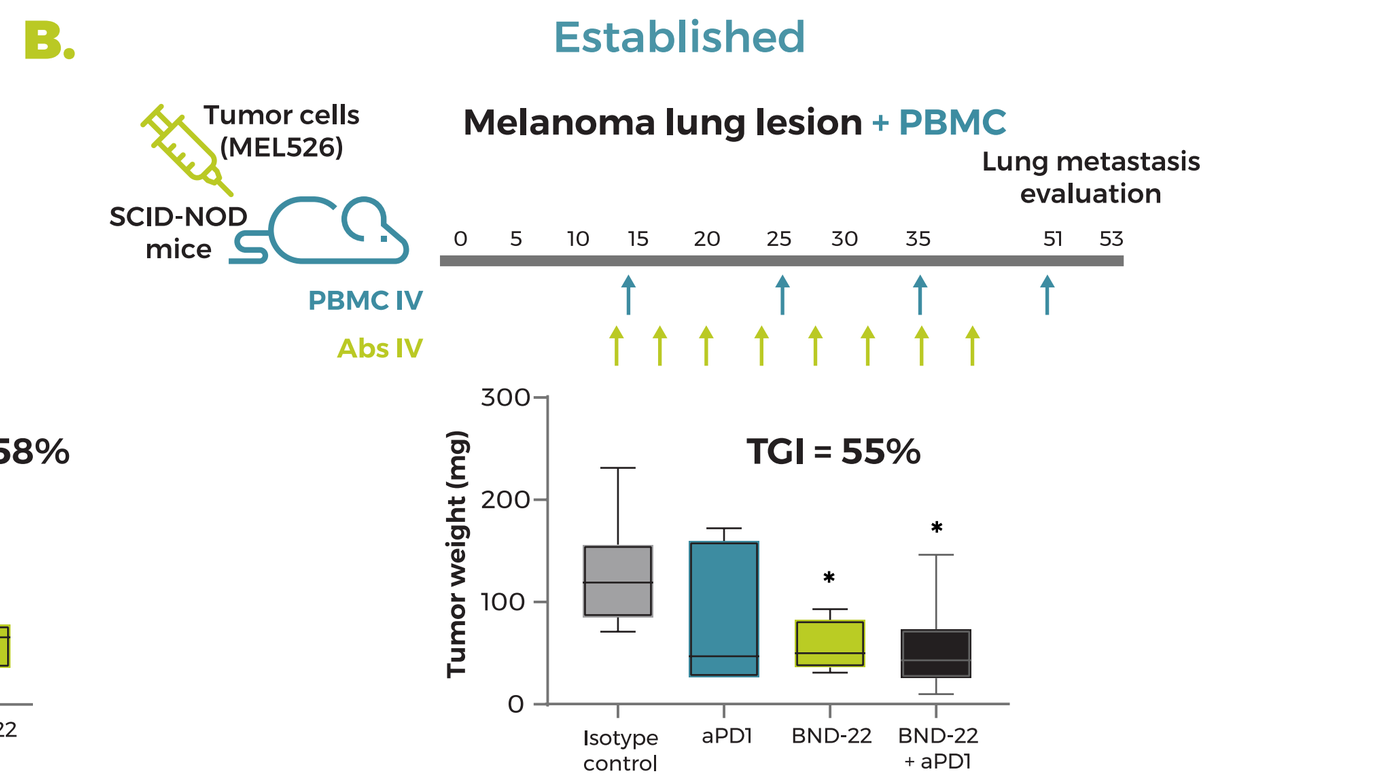
Tumor cells (MEL526)
SCID-NOD mice
Melanoma lung lesion + PBMC
Lung metastasis evaluation
PBMC IV
Abs IV
Tumor weight (mg)
TGI = 55%
Isotype control
aPD1
BND-22
BND-22 + aPD1

C. BND-22 enhances macrophage anti-tumor activity: in vivo colon cancer model

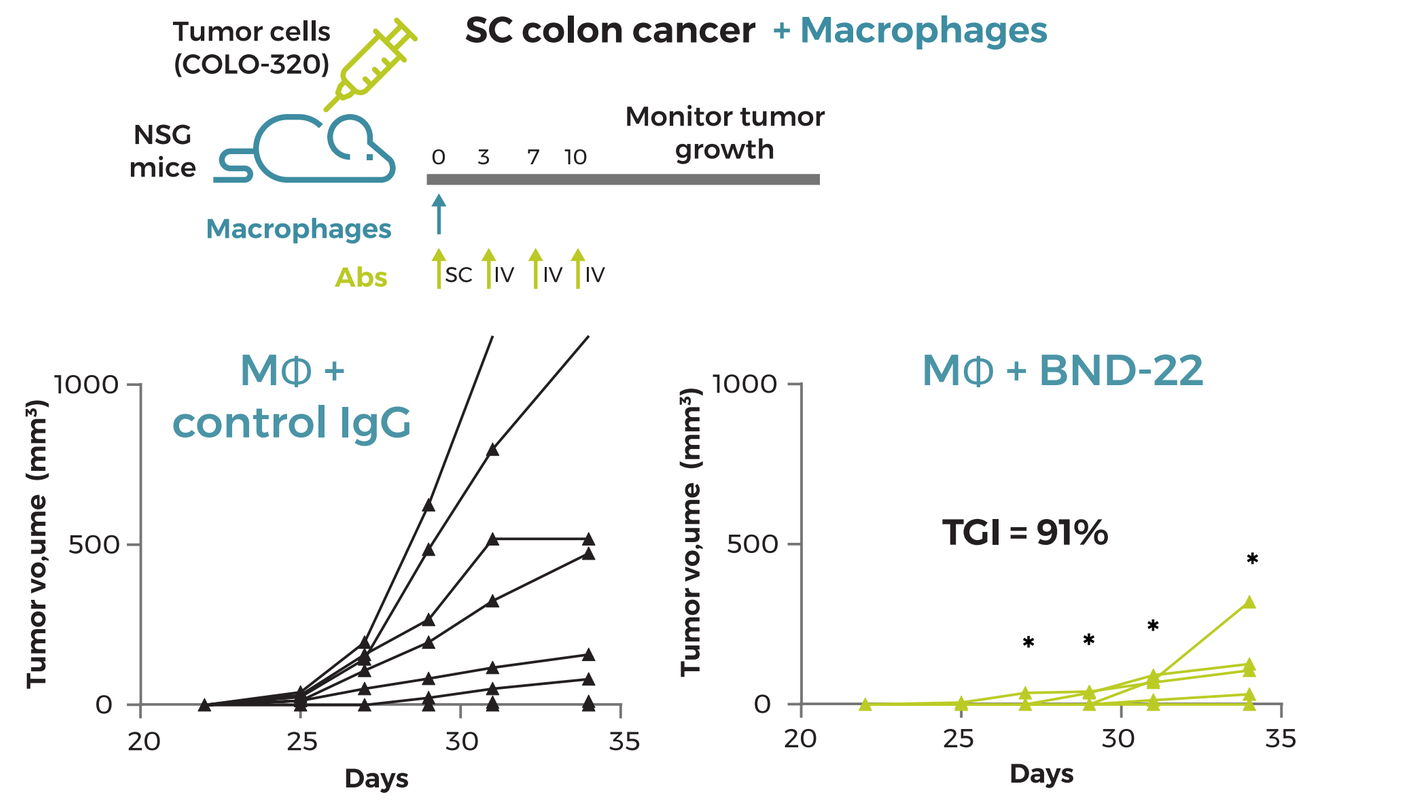
Tumor cells (COLO-320)
NSG mice
SC colon cancer + Macrophages
Monitor tumor growth
Macrophages
Abs
MΦ + control IgG
MΦ + BND-22
TGI = 91%
Tumor volume (mm³)
Days

D. BND-22 enhances anti-tumor immune activity: in vivo humanized H&N cancer model

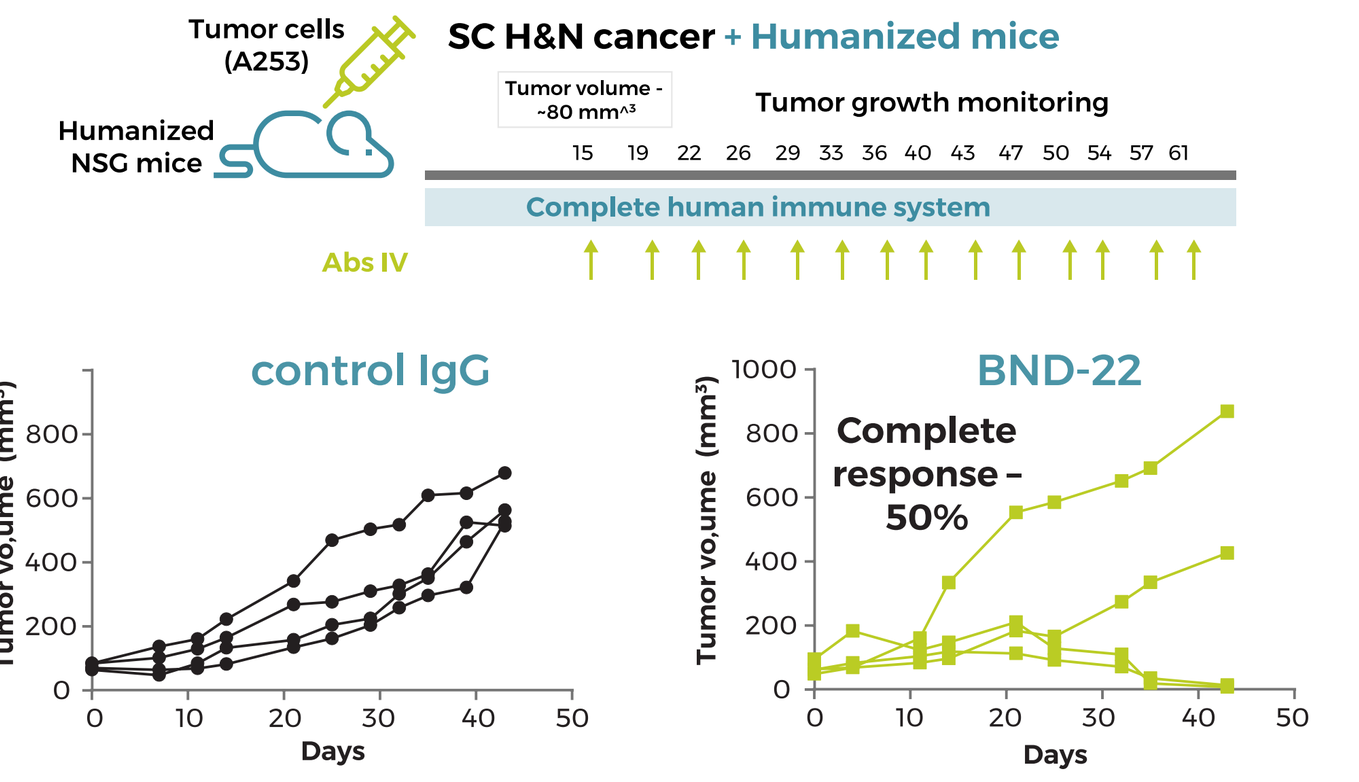
Tumor cells (A253)
Humanized NSG mice
SC H&N cancer + Humanized mice
Tumor volume - ~80 mm³
Tumor growth monitoring
Complete human immune system
Abs IV
control IgG
BND-22
Complete response – 50%
Tumor volume (mm³)
Days

A,B. SCID-NOD mice were engrafted IV with MEL526 cells. Human PBMC isolated from healthy donors were administrated to the mice together with the relevant treatments starting form day 1 (A) or day 14 (B) according to the treatment schedules indicated in the illustrations. Images demonstrating the lesions generated by the melanoma cells and the tumor weight in the mice' lungs from the different groups are presented (A,B). C. NSG mice were engrafted SC with COLO-320 cells and human macrophages generated from healthy donors together with the indicated treatments. D. Humanized human CD34+-engrafted mice were inoculated with A253 cells. The mice were treated according to the indicated treatment schedule when tumors reached 80mm³. Student's T test was used to calculate statistical significance in tumor growth inhibition (TGI) between the mean tumor volume or weight in the various experiments (A-C) * P <0.05

10 ILT2 pathway-related predictive biomarkers will be explored in clinical settings

Biomarkers strategy was established according to the MOA of BND-22

Biomarker	Method
Membranal HLA-G	IHC, Real-time PCR
Soluble HLA-G	ELISA, Real-time PCR
ILT2 expression by tumor infiltrating immune cells	IHC
ILT2 expression by peripheral immune cells	FACS
Expression of pro-phagocytic molecules by tumor cells	FACS
Immune cell infiltration	IHC/FACS

The effect of BND-22 is correlated with the expression of ILT2 in different experimental systems

A.

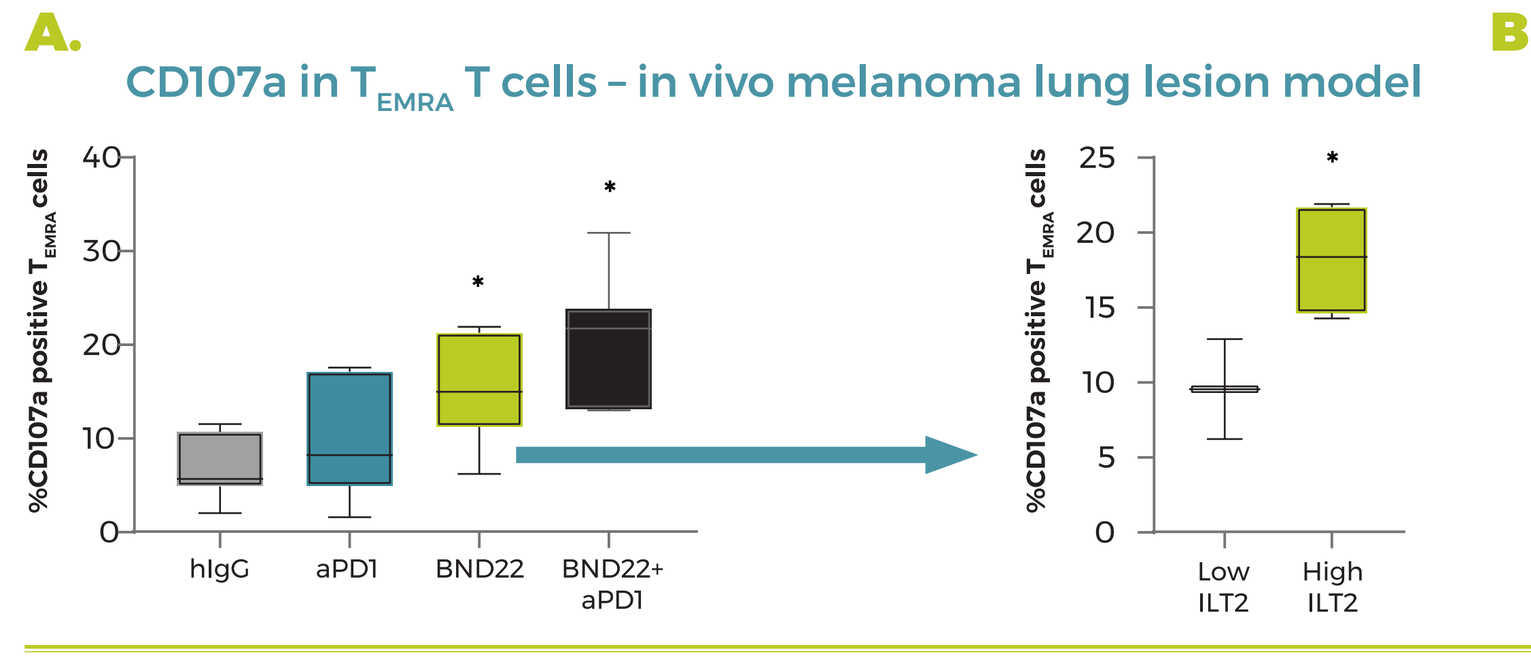
CD107a in TEMRA T cells – in vivo melanoma lung lesion model
%CD107a positive TEMRA cells
hIgG
aPD1
BND22
BND22+ aPD1
Low ILT2
High ILT2

B.

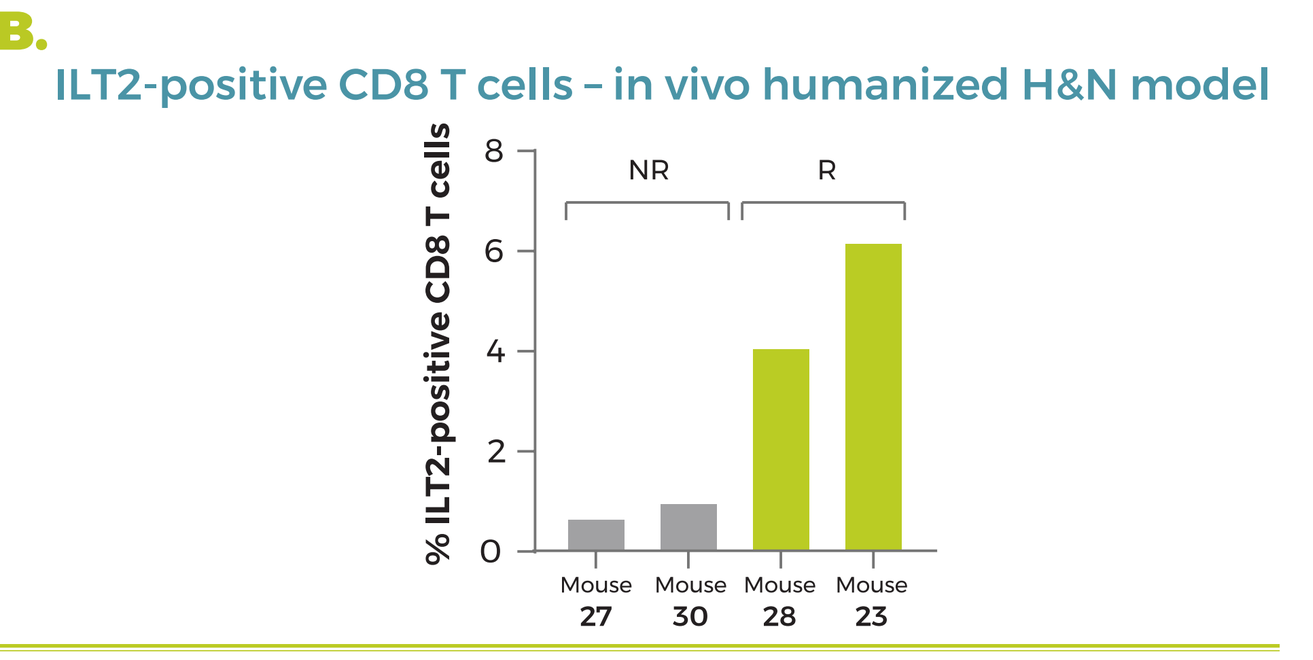
ILT2-positive CD8 T cells – in vivo humanized H&N model
% ILT2-positive CD8 T cells
NR
R
Mouse 27
Mouse 30
Mouse 28
Mouse 23

A. The levels of CD107a in T_{EMRA} T cells were evaluated in mice from the lung lesion melanoma established model (left). The levels of ILT2 in the TEMRA T cells from the different donors and effect (% of CD107a -positive TEMRA T cells). (middle). B. The levels of ILT2-positive CD8 T cells were evaluated in humanized mice inoculated with H&N cancer that responded (R) or did not respond (NR) to BND-22.

11 Summary

- BND-22 is a first-in class, ILT2-targeting, multi-cell checkpoint inhibitor
- Preclinical program has demonstrated that BND-22 enhances the anti-tumor activity of innate and adaptive immune cells in different systems, including patient-derived tumor samples and various in-vivo models
- ILT2 multi-cell effect and regulation offers a differentiated MoA, including potential benefit for anti-PD-1 non-responders or relapsing patients
- GLP Toxicity study in Cynomolgus monkeys completed with no mortality, clinical signs or gross pathology findings
- Scientific data and preclinical studies support studying BND-22 as a single agent in clinical trials
- Expression of HLA-G in tumor cells and ILT2 in immune cells are potential biomarkers for predicting response to BND-22
- Safety, tolerability, and anti-tumor activity of BND-22 will be studied in a first-in-human clinical trial (Q1 2021)