CD28 shedding is a novel immune-regulatory mechanism found in cancer patients which directly inhibits anti PD-1 effect

Authors: Motti Hakim[1], Anna Fridman Dror[1], Dror Alishekevitz[1], Edna Meilin[1], Orit Shilovizky[1], Jeffrey S Weber[2], Yair Sapir[1], Avidor Shulman[1], Tehila Ben Moshe[1]. **Affiliation:** (1) Biond Biologics, Misgav industrial park, Israel. (2) Perlmutter Cancer Center, NYU Langone Health, New York, USA.

Visit our website

Poster Session Title:
Tumor-immune System Interactions 2
Poster Number: 2846A

1 Background

Background

The interaction between the CD28 receptor and its B7 ligands has a crucial role in the induction of a potent and persistent anti-tumor T cell response. Indeed, the most effective immune oncology treatments target CTLA-4 and PD-1, proteins that directly downregulate CD28 activity, further emphasizing the need for better understanding of the regulation of CD28.

Shedding of immune receptors is one mechanism that the immune system employs to modulate signalling cascades. Here, we describe, for the first time, the finding that membranal CD28 is actively shed by specific proteases upon T cell stimulation in humans, and its implications on immune oncology.

Results

Elevated levels of soluble CD28 were found in 15% of plasma samples from cancer patients across various tumor types. A variety of T cell stimulations in vitro resulted in release and accumulation of soluble CD28. The origin of soluble CD28 can be ascribed to proteolytic cleavage of the receptor, as treatments with protease inhibitors diminished the amount of soluble CD28 in a concentration-dependent manner. Moreover, the shedding of membranal CD28 is found to be a continuous process even after removal of the stimuli. The proteolytic site was mapped to the stalk region of human CD28. In various MLR assays, the presence of soluble CD28 resulted in inhibition of effector cytokine secretion and T cells clustering together with elevation in tumor supportive cytokines, such as IL-6, IL-10 and IDO. Importantly, soluble CD28 counteracted anti PD-1 activity and inhibited the secretion of inflammatory cytokines, such as IL-2 and IFNγ. Evaluation of plasma samples from patients undergoing anti-PD1 therapy demonstrated an association of clinical response with decreased plasma levels of soluble CD28.

Conclusions

We report here the discovery of a novel regulatory mechanism which blocks the CD28/B7 activation pathway by generating a decoy human CD28 receptor. This regulatory mechanism occurs in the setting of cancer as aberrant levels of soluble CD28 were found in the plasma of cancer patients, where its presence was associated with reduced response to anti-PD1 therapy. The development of therapeutic drug candidates for the inhibition of this regulatory mechanism is in progress.

2 Aberrant levels of soluble-CD28 were found in plasma samples of cancer patients

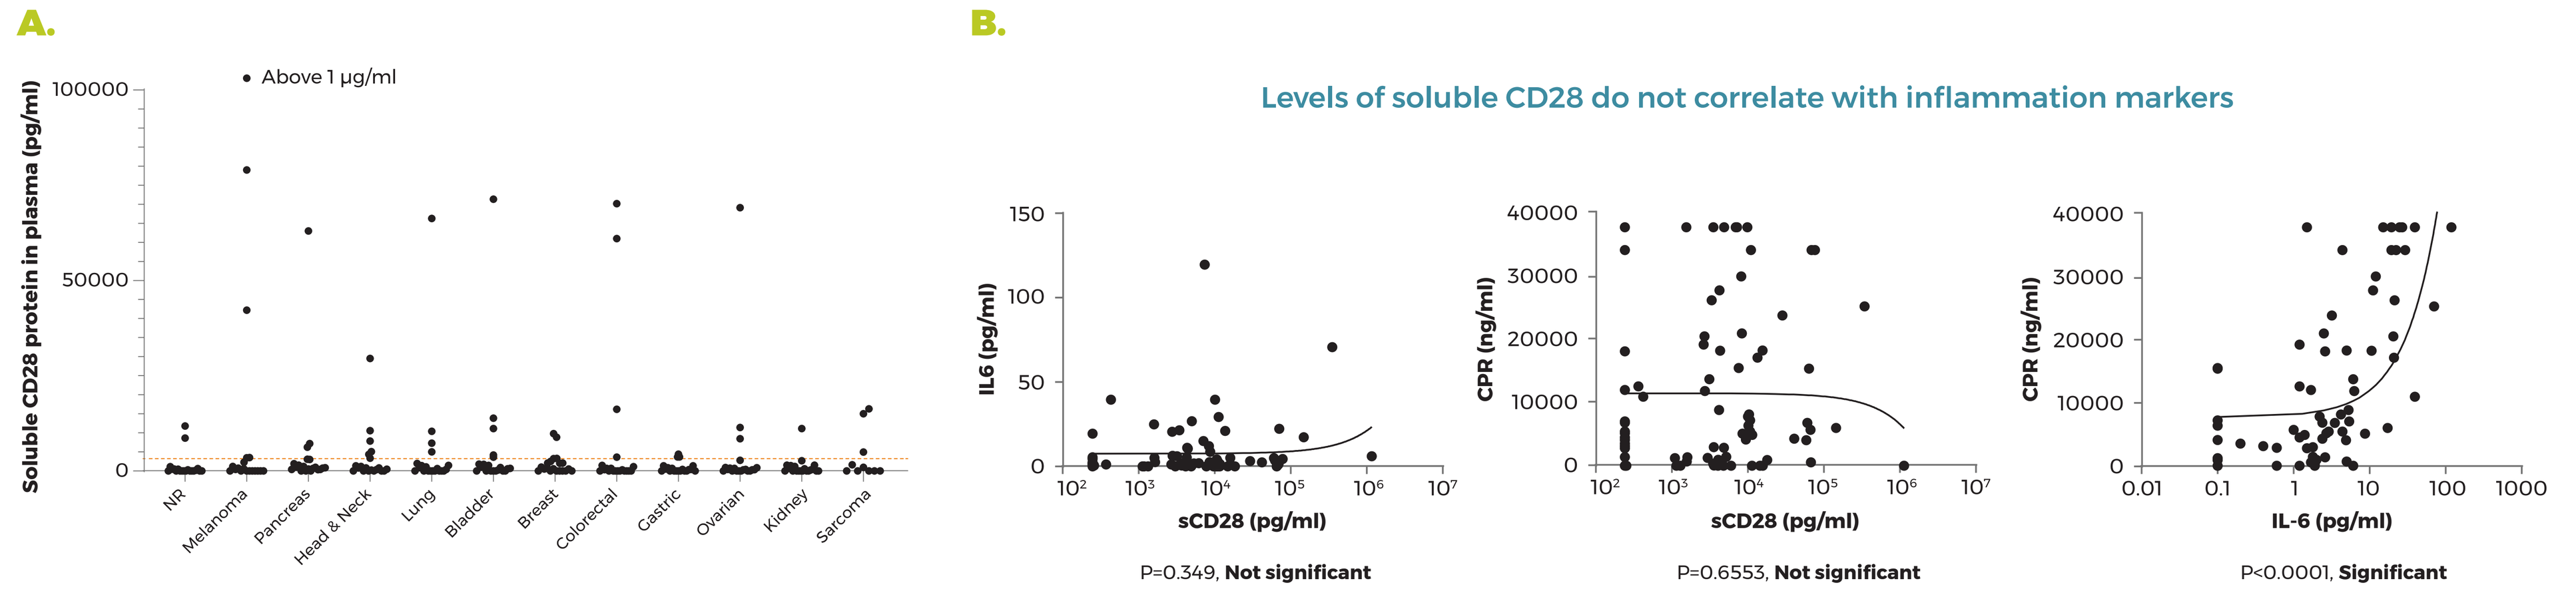
A.
Above 1 µg/ml
Soluble CD28 protein in plasma (pg/ml)
NR
Melanoma
Pancreas
Head & Neck
Lung
Bladder
Breast
Colorectal
Gastric
Ovarian
Kidney
Sarcoma
B.
Levels of soluble CD28 do not correlate with inflammation markers
IL6 (pg/ml)
sCD28 (pg/ml)
P=0.349, Not significant
CPR (ng/ml)
sCD28 (pg/ml)
P=0.6553, Not significant
CPR (ng/ml)
IL-6 (pg/ml)
P<0.0001, Significant

Plasma samples of eleven tumor types were quantified for amount of soluble CD28 by a proprietary ELISA and compared to samples derived from healthy subjects (A). Red line indicates soluble CD28 threshold which is 2.5 fold above the reported amount for healthy subjects. NR- healthy subjects. 40 plasma samples with low to high soluble CD28 levels were assayed for CRP and IL-6 levels with specialized ELISA (B). Correlation tests were performed using GraphPad by utilizing regression fit calculations.

3 The CD28 receptor was found to be proteolytically cleaved in activated T cells

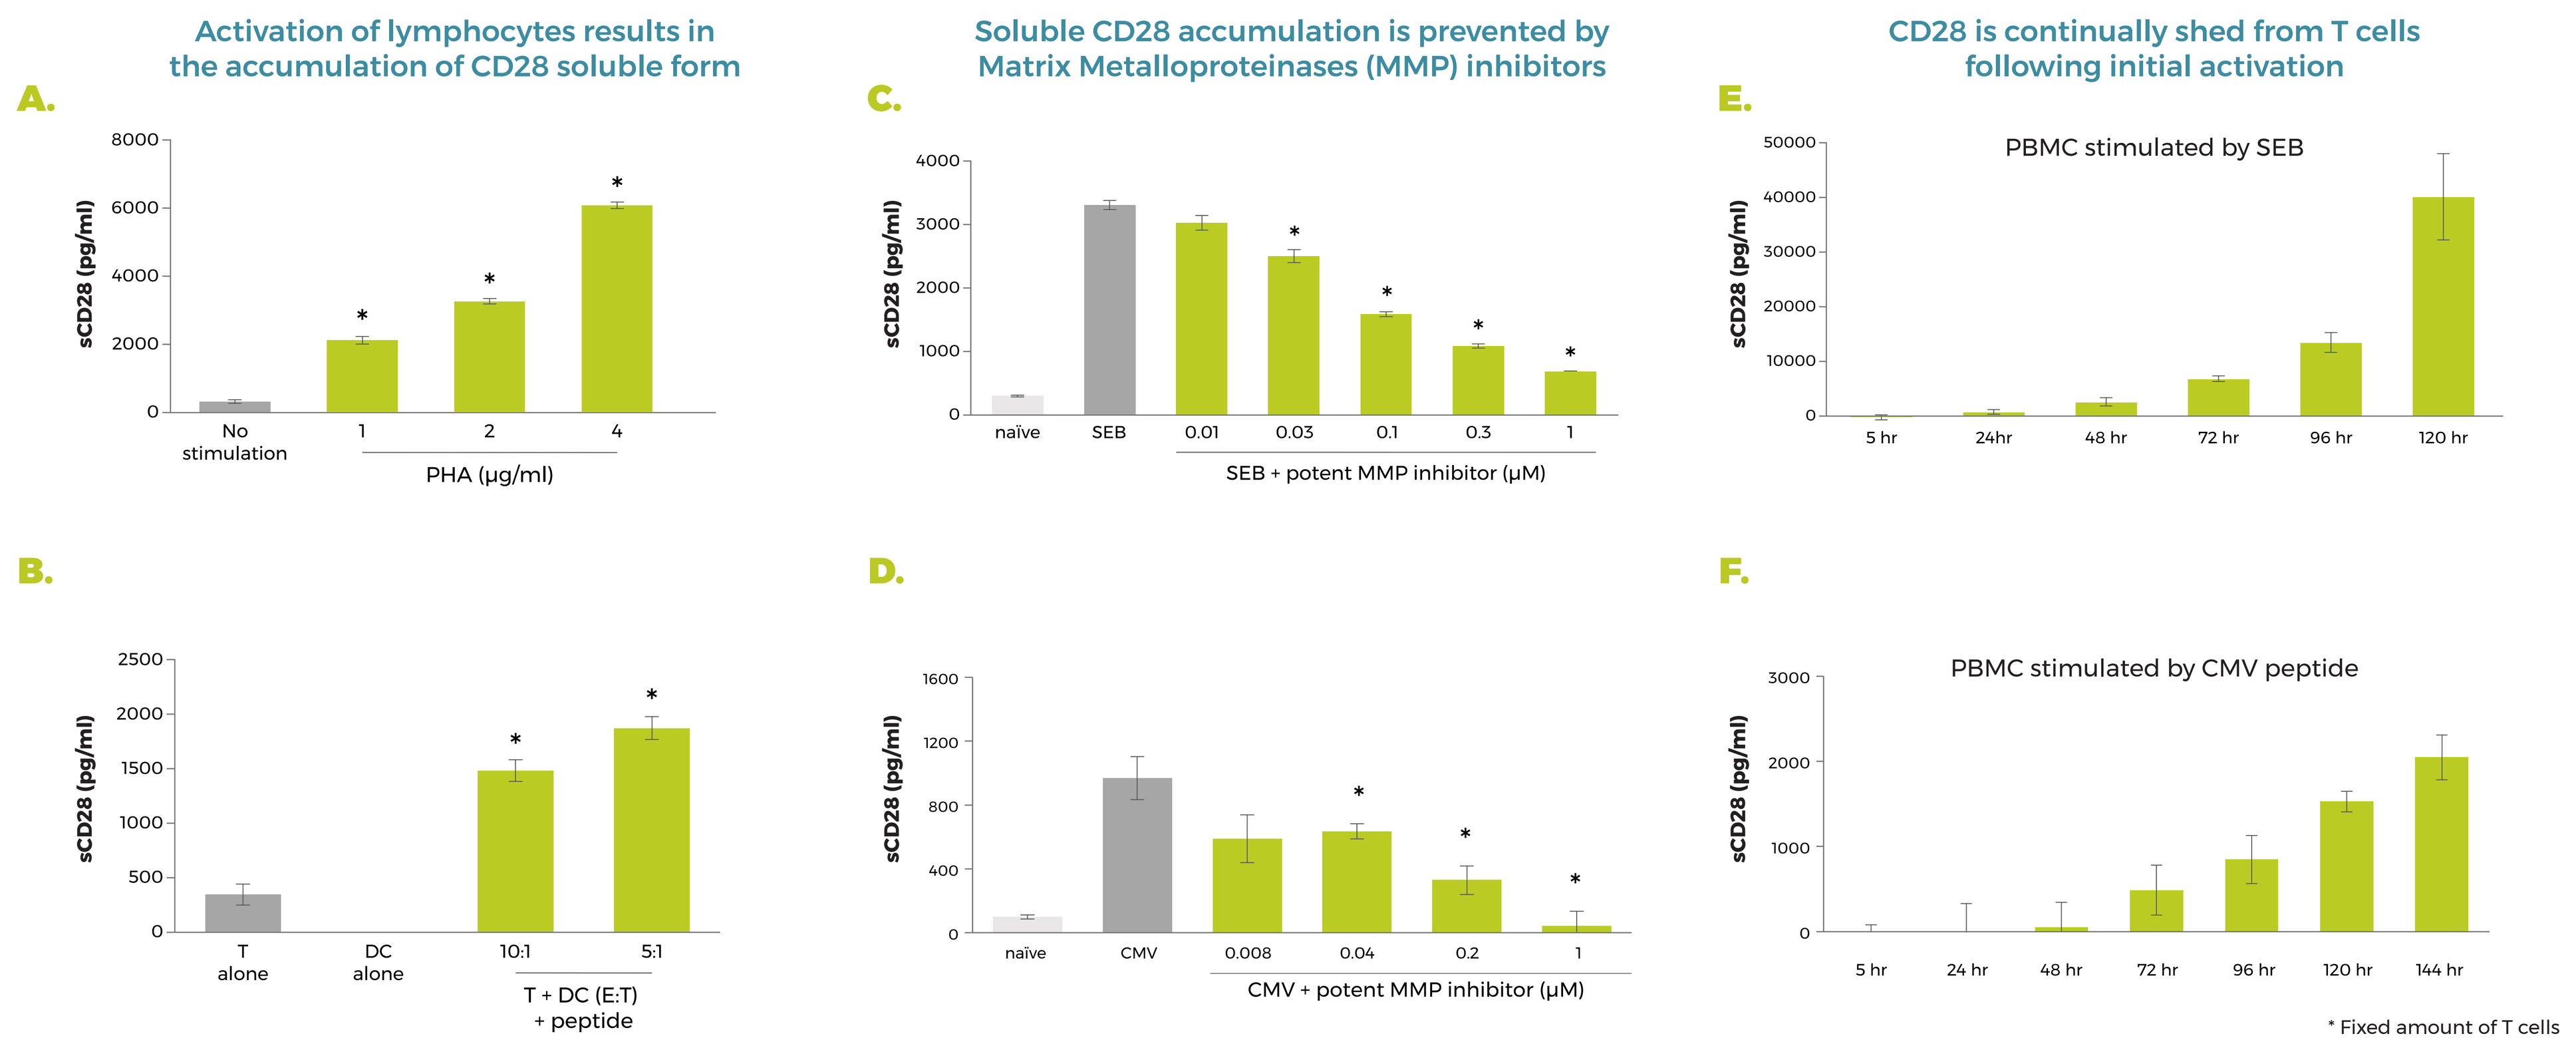
Activation of lymphocytes results in the accumulation of CD28 soluble form
A.
sCD28 (pg/ml)
No stimulation
PHA (µg/ml)
B.
T alone
DC alone
T + DC (E:T) + peptide
Soluble CD28 accumulation is prevented by Matrix Metalloproteinases (MMP) inhibitors
C.
naïve
SEB
SEB + potent MMP inhibitor (µM)
D.
CMV
CMV + potent MMP inhibitor (µM)
CD28 is continually shed from T cells following initial activation
E.
PBMC stimulated by SEB
F.
PBMC stimulated by CMV peptide
* Fixed amount of T cells

Soluble CD28 (sCD28) is found in culture media of isolated T cells stimulated by PHA (A) or autologous dendritic cells presenting CMV antigen peptides (B). Soluble CD28 accumulation is mediated by MMPs as treatments with a potent inhibitor of MMPs reduces the amount of detected soluble CD28 in culture media of PBMC stimulated by Staphylococcal enterotoxin B (SEB, C) or Cytomegalovirus (CMV) peptides (D) in a dose dependent manner. CD28 is continually shed from PBMC that were stimulated with low amount of SEB (E) or CMV peptides (F) for a short period of time, washed extensively and incubated without stimulant for the indicated time. Soluble CD28 was detected using proprietary in-house ELISA. MMP inhibitor is TMI-1 (GI254023X was also used with same effect). P<0.05, unpaired student T.test compared to no stimulation or T cells alone.

4 Model describing the immuno-suppressive effect of CD28 shedding

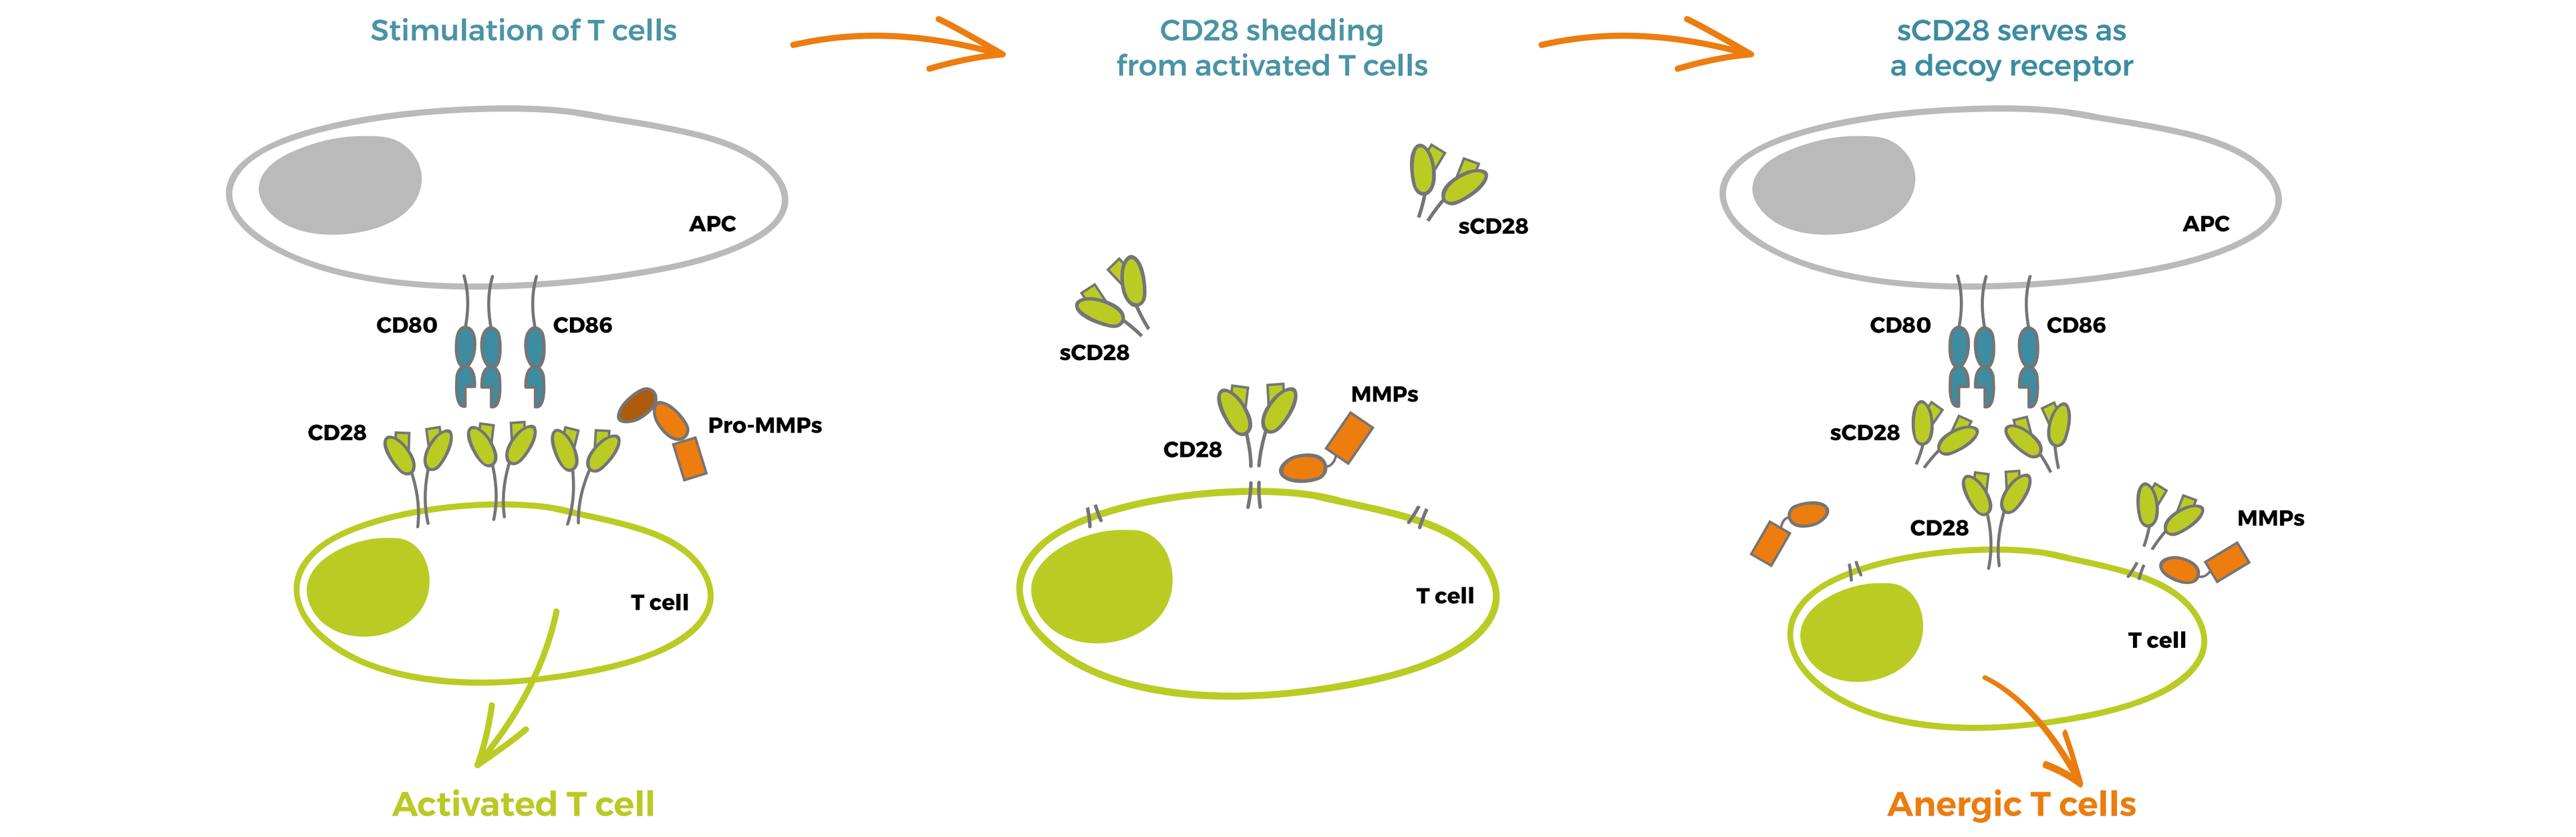
Stimulation of T cells
CD28 shedding from activated T cells
sCD28 serves as a decoy receptor
APC
CD80
CD86
CD28
Pro-MMPs
T cell
Activated T cell
sCD28
MMPs
Anergic T cells

During the activation of T cells, Matrix MetalloProteinases (MMPs) are upregulated (left panel) and fully activated by pro-domain removal. MMPs cleave CD28 at its stalk region (center panel), releasing a dimeric soluble CD28 that acts as a decoy receptor by binding to CD28 cognate ligands, CD80 and CD86 (B7 molecules) on antigen presenting cells (APC). Tumor residing T cells with lower membranal CD28 density and insufficient B7 mediated signaling will fall into anergic state with low capacity to be rescued by anti-PD1 therapy (right panel).

5 MMP-2 and MMP-13 were found to cleave human CD28 at its stalk region between P144 and L145

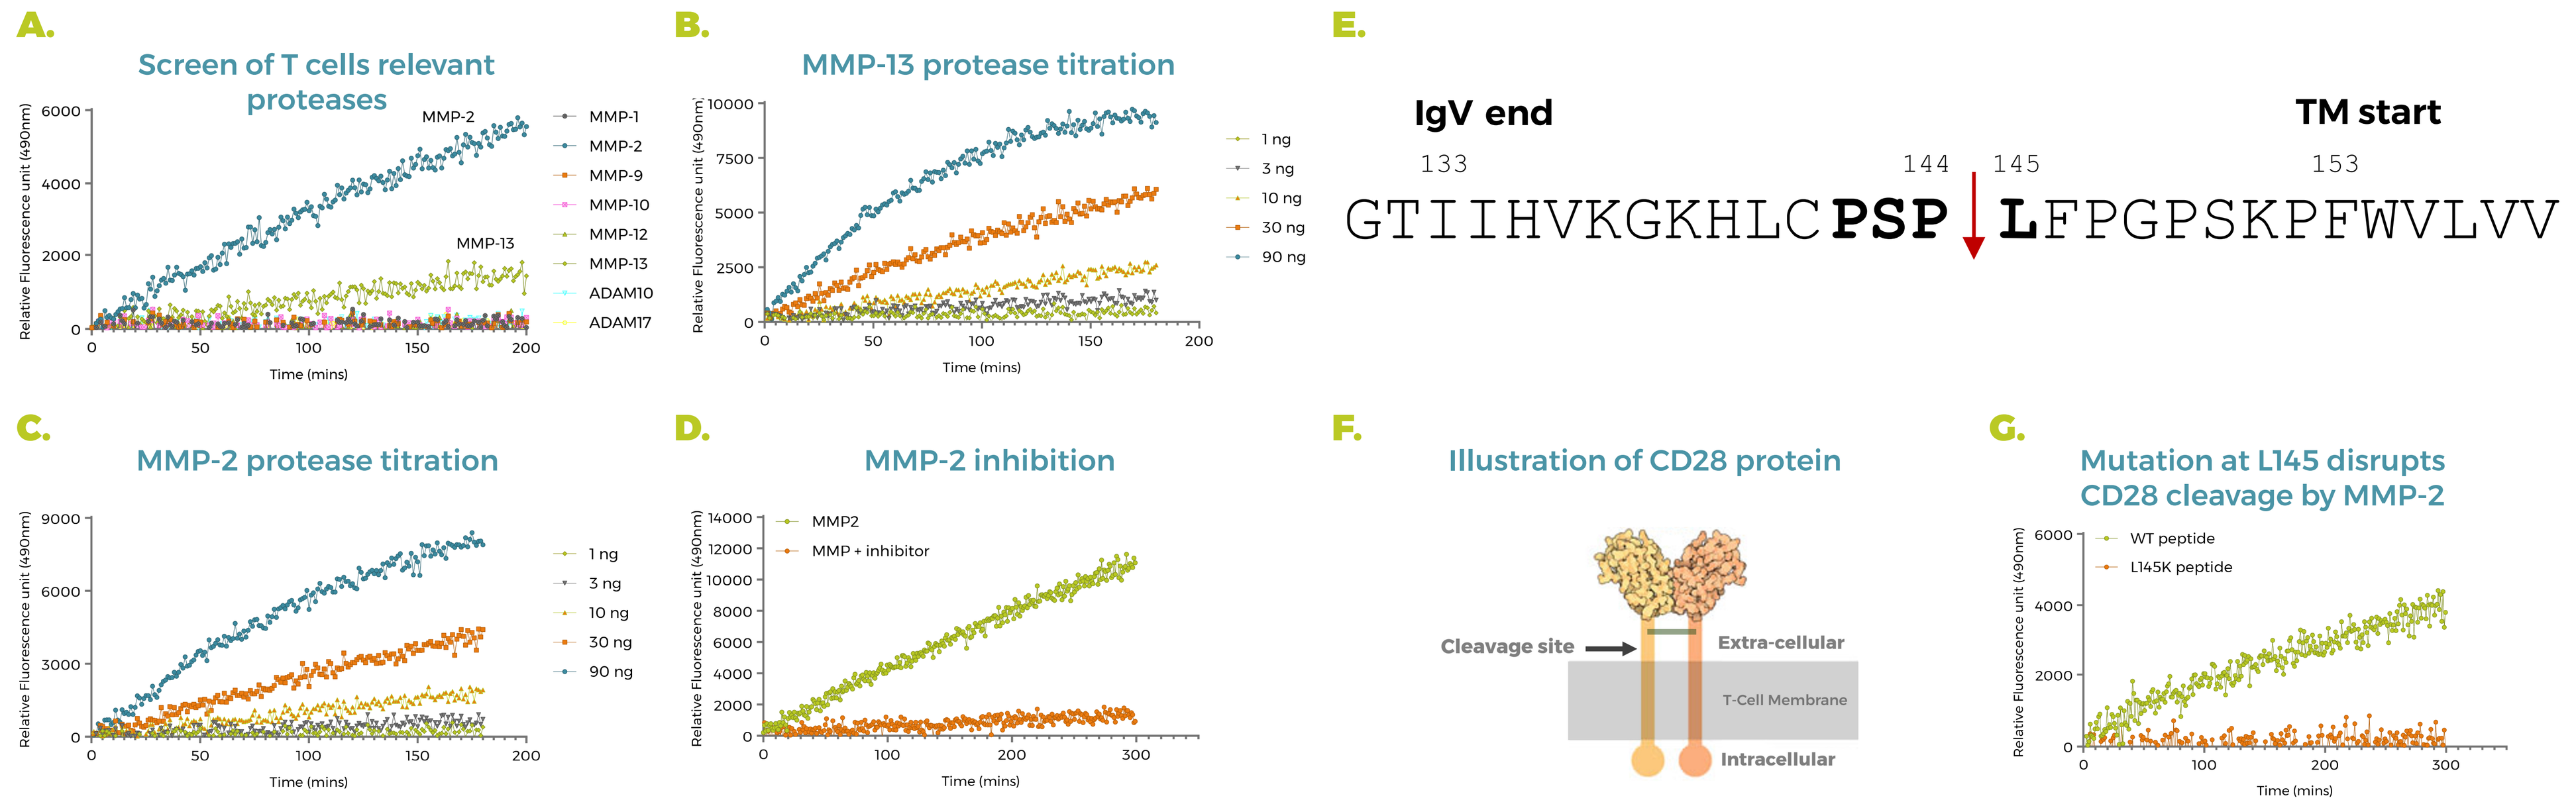
A.
Screen of T cells relevant proteases
MMP-1
MMP-2
MMP-9
MMP-10
MMP-12
MMP-13
ADAM10
ADAM17
Time (mins)
B.
MMP-13 protease titration
1 ng
3 ng
10 ng
30 ng
90 ng
C.
MMP-2 protease titration
D.
MMP-2 inhibition
MMP2
MMP + inhibitor
E.
IgV end
TM start
133
144
145
153
GTIIHVKGKHLCPSP LFPGPSKPFWVLVV
F.
Illustration of CD28 protein
Cleavage site
Extra-cellular
T-Cell Membrane
Intracellular
G.
Mutation at L145 disrupts CD28 cleavage by MMP-2
WT peptide
L145K peptide

Screen of different MMPs and ADAM protein family members identified MMP-2 and MMP-13 with proteolytic activity toward a quenched fluorogenic peptide corresponding to human CD28 stalk region (A). Cleavage activity was further validated by titration of MMP-13 (B), MMP-2 (C) and specific inhibition (100nM TMI-1) of MMP-2 (D) or MMP-13 (data not shown). Location of ectodomain cleavage site in human CD28 was Identified by mass spectrometry to reside between Pro144 and Leu145, a location that is part of a PXX/Xhyd sequence, a known consensus motif for MMP substrates (E). The location of the cleavage site relative to CD28 homo-dimeric structure is illustrated (F). Validation of cleavage site was done by mutating Leu145 to Lysine and subjecting it to MMP-2 (G).

6 Soluble CD28 promotes an immuno-suppressive environment

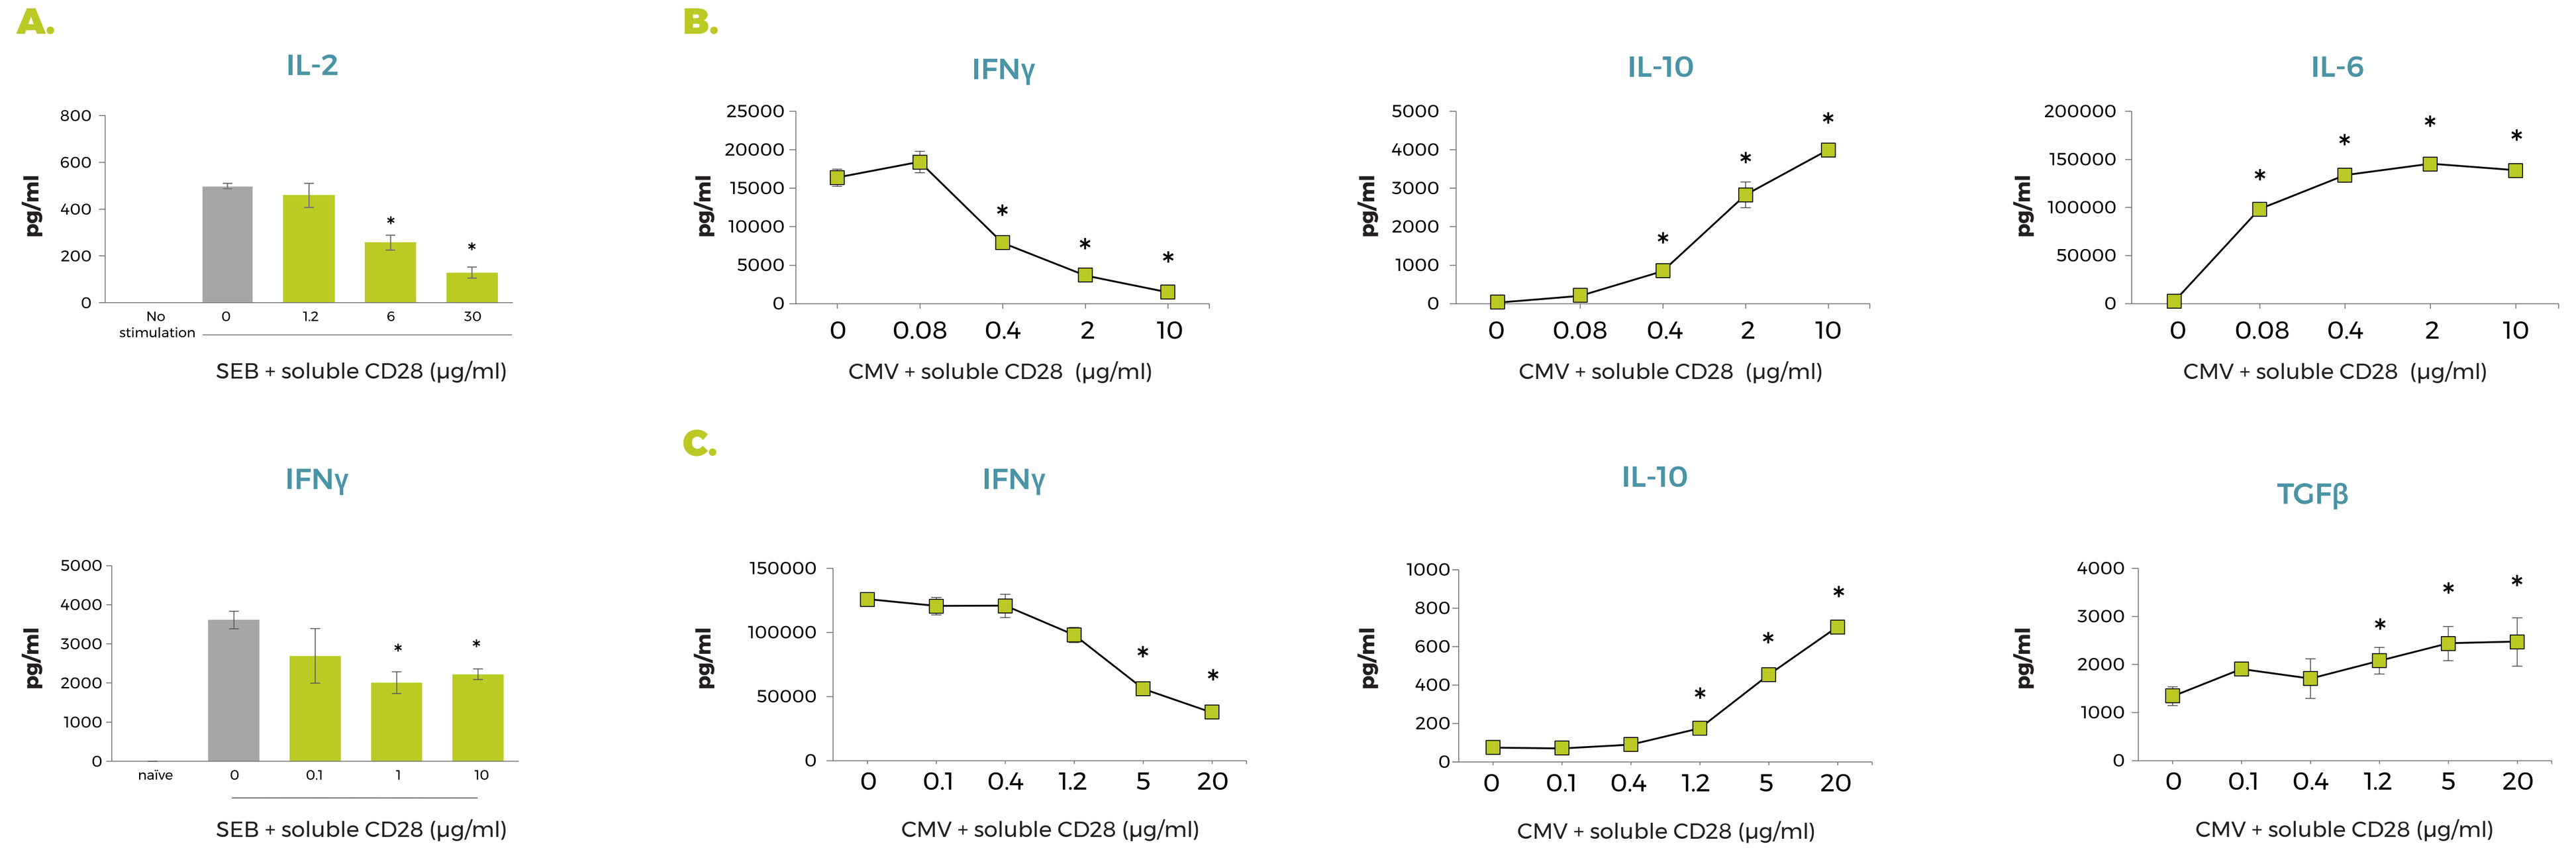
A.
IL-2
pg/ml
No stimulation
SEB + soluble CD28 (µg/ml)
IFNγ
naïve
B.
IFNγ
IL-10
IL-6
CMV + soluble CD28 (µg/ml)
C.
IFNγ
IL-10
TGFβ
CMV + soluble CD28 (µg/ml)

Soluble CD28 creates a tumor supportive environment evident by diminished effector cytokine secretion (IFNγ and IL-2) and elevation of suppressive immune-molecules (IL-10 and TGFβ) as demonstrated by PBMC stimulated with SEB superantigen (A) or CMV peptides (B) or by autologous monocytes and CD3 T cells MLR (C). Cytokine levels were measured by ELISA. * P<0.05, unpaired student's t-test compared to stimulation only.

7 Soluble CD28 attenuates anti-PD-1 immunostimulatory effect

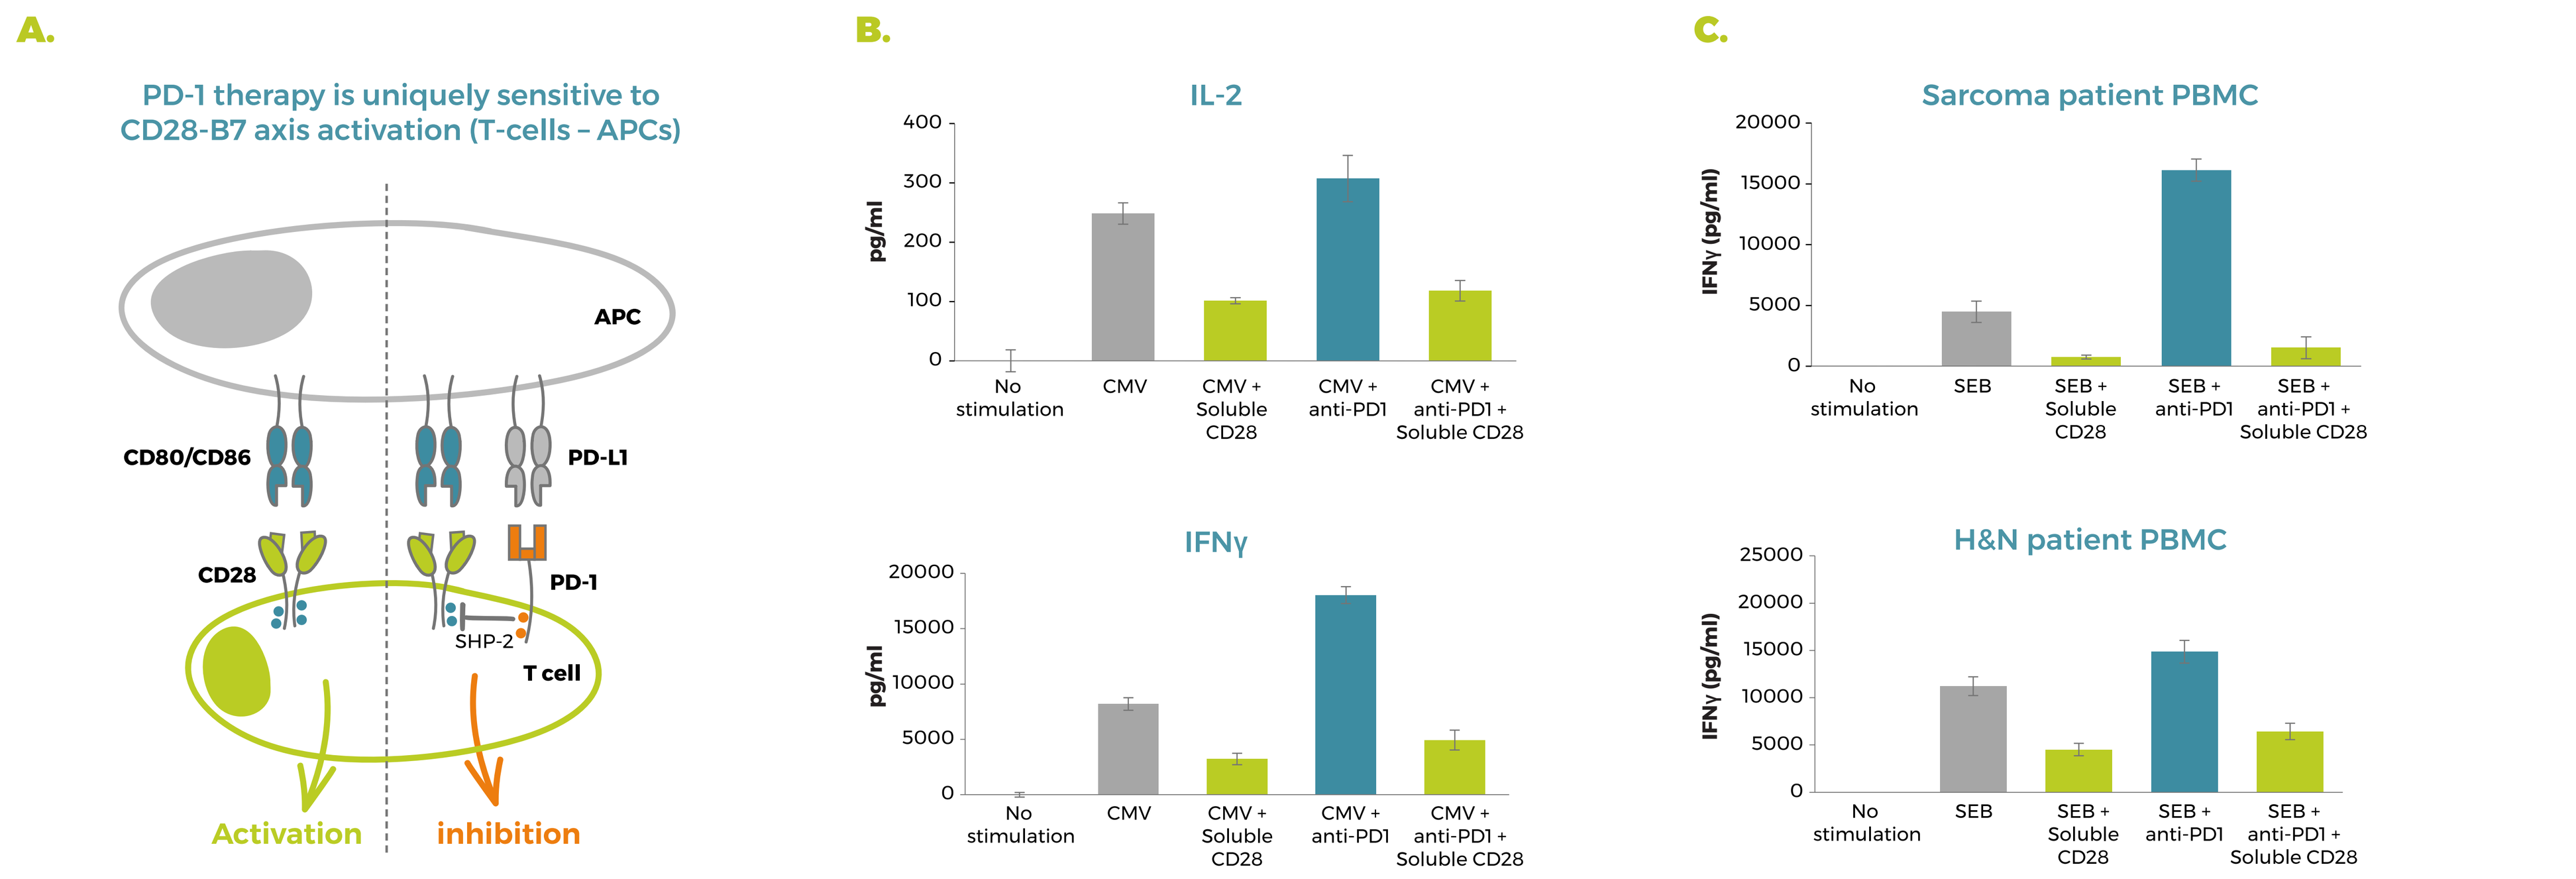
A.
PD-1 therapy is uniquely sensitive to CD28-B7 axis activation (T-cells – APCs)
APC
CD80/CD86
PD-L1
CD28
PD-1
SHP-2
T cell
Activation
inhibition
B.
IL-2
IFNγ
pg/ml
No stimulation
CMV
CMV + Soluble CD28
CMV + anti-PD1
CMV + anti-PD1 + Soluble CD28
C.
Sarcoma patient PBMC
H&N patient PBMC
IFNγ (pg/ml)
SEB
SEB + Soluble CD28
SEB + anti-PD1
SEB + anti-PD1 + Soluble CD28

PD-1 inhibition is mediated by reducing tyrosine phosphorylation specifically on CD28 ITAM sequences as illustrated in (A) and described by Hui E, Science, 2017. Hence, the effect of PD-1 blockade is dependent on intact CD28 signaling as documented by Kamphorst AO, Science, 2017. Blocking CD28/B7 axis by soluble CD28 molecules counteracts the positive effect of anti-PD1 (Keytruda, blue bars) on effector cytokine secretion (IFNγ and IL-2) of healthy donor PBMC stimulated by CMV (B) and cancer patients' PBMC stimulated by SEB (C). Cytokine levels were measured by ELISA. * P<0.05, unpaired student's t-test compared to anti-PD1 treatment alone.

8 Soluble CD28 dynamics relates with response to PD-1 pathway therapy

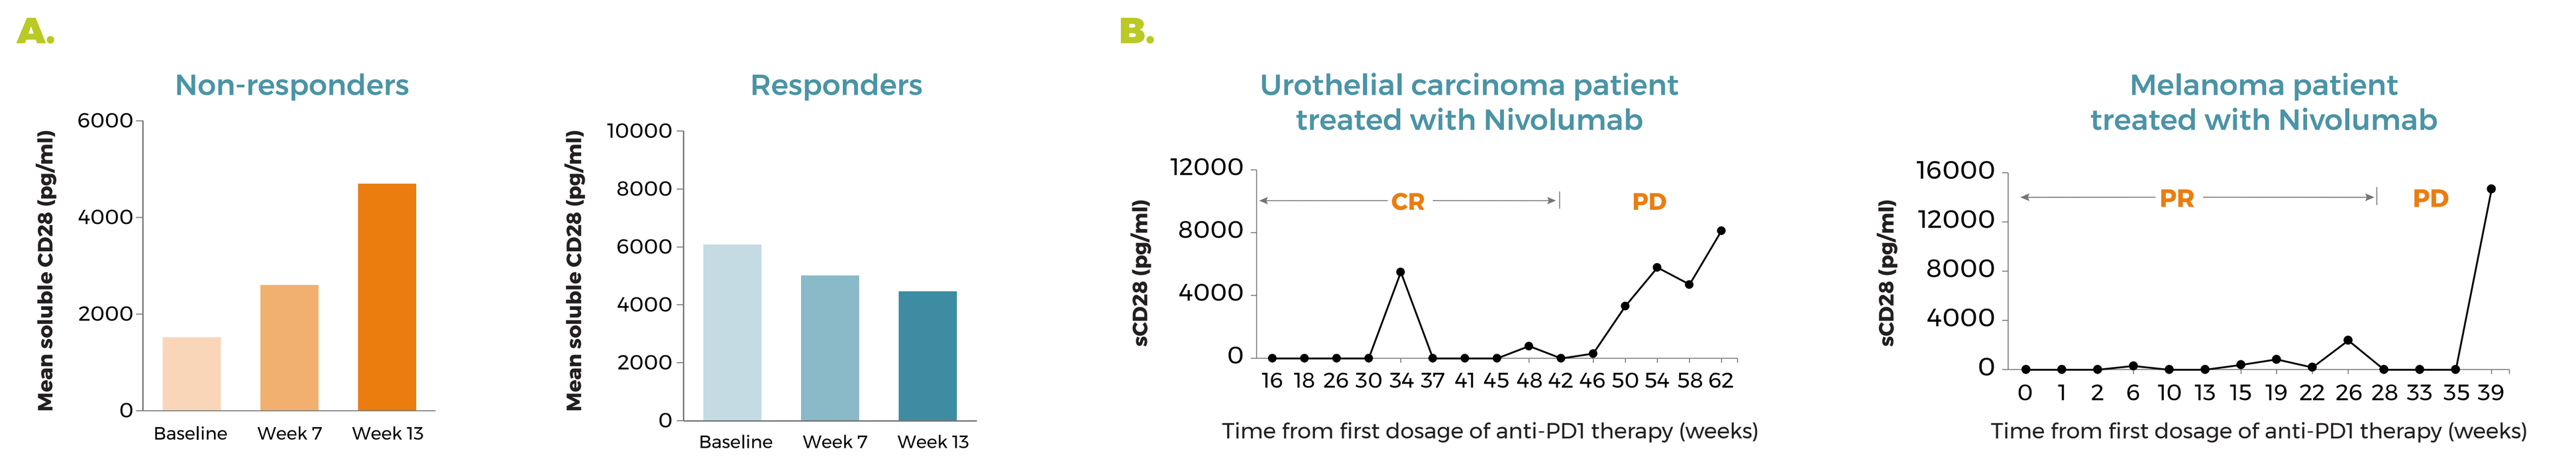
A.
Non-responders
Responders
Mean soluble CD28 (pg/ml)
Baseline
Week 7
Week 13
B.
Urothelial carcinoma patient treated with Nivolumab
Melanoma patient treated with Nivolumab
sCD28 (pg/ml)
CR
PD
PR
Time from first dosage of anti-PD1 therapy (weeks)

A) Average serum levels of soluble CD28 in melanoma patients during anti-PD1 therapy stratified according to response. (Non-responders, N=47; Responders, N=46). (B) Two case studies hints to the resistance mechanism to anti-PD1 therapy invoked by accumulation of soluble CD28. Soluble CD28 was detected using proprietary in-house ELISA. PD – progressed disease; PR- partial response; CR – complete response.

9 BION-206 - An efficient VHH antibody that blocks the shedding of CD28 receptor

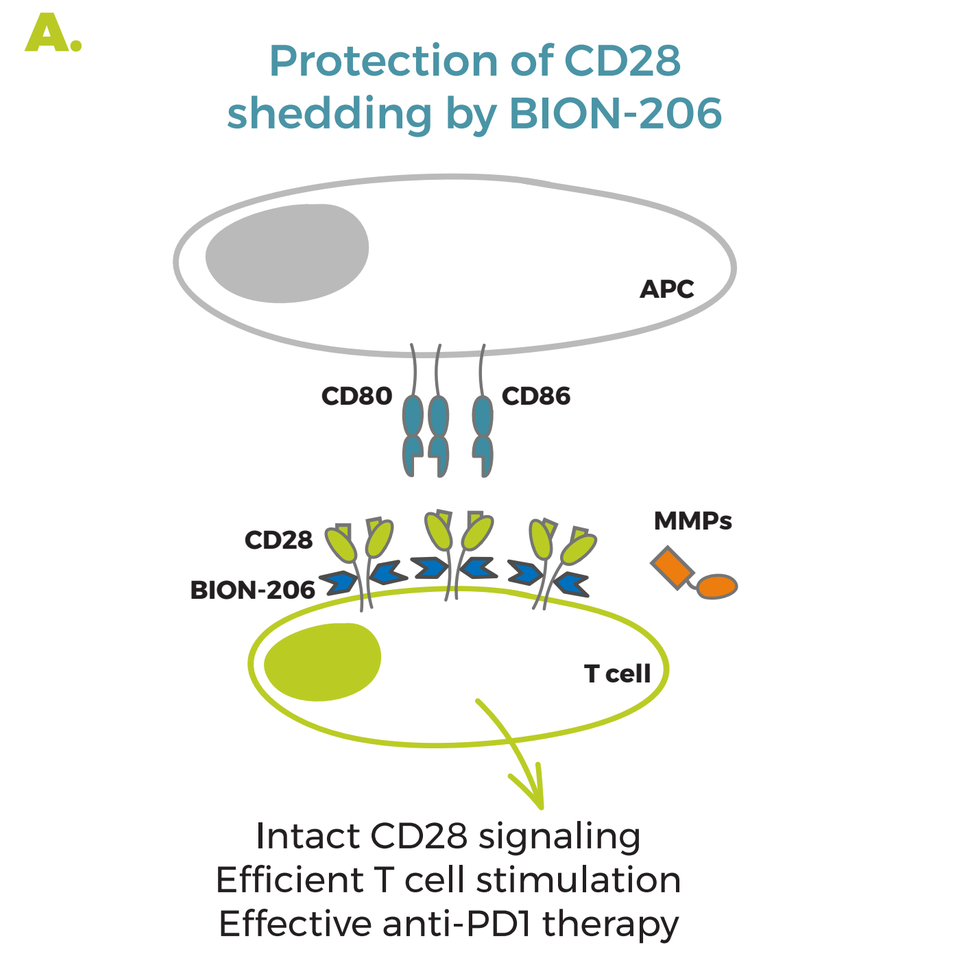
A.
Protection of CD28 shedding by BION-206
APC
CD80
CD86
CD28
BION-206
MMPs
T cell
Intact CD28 signaling
Efficient T cell stimulation
Effective anti-PD1 therapy

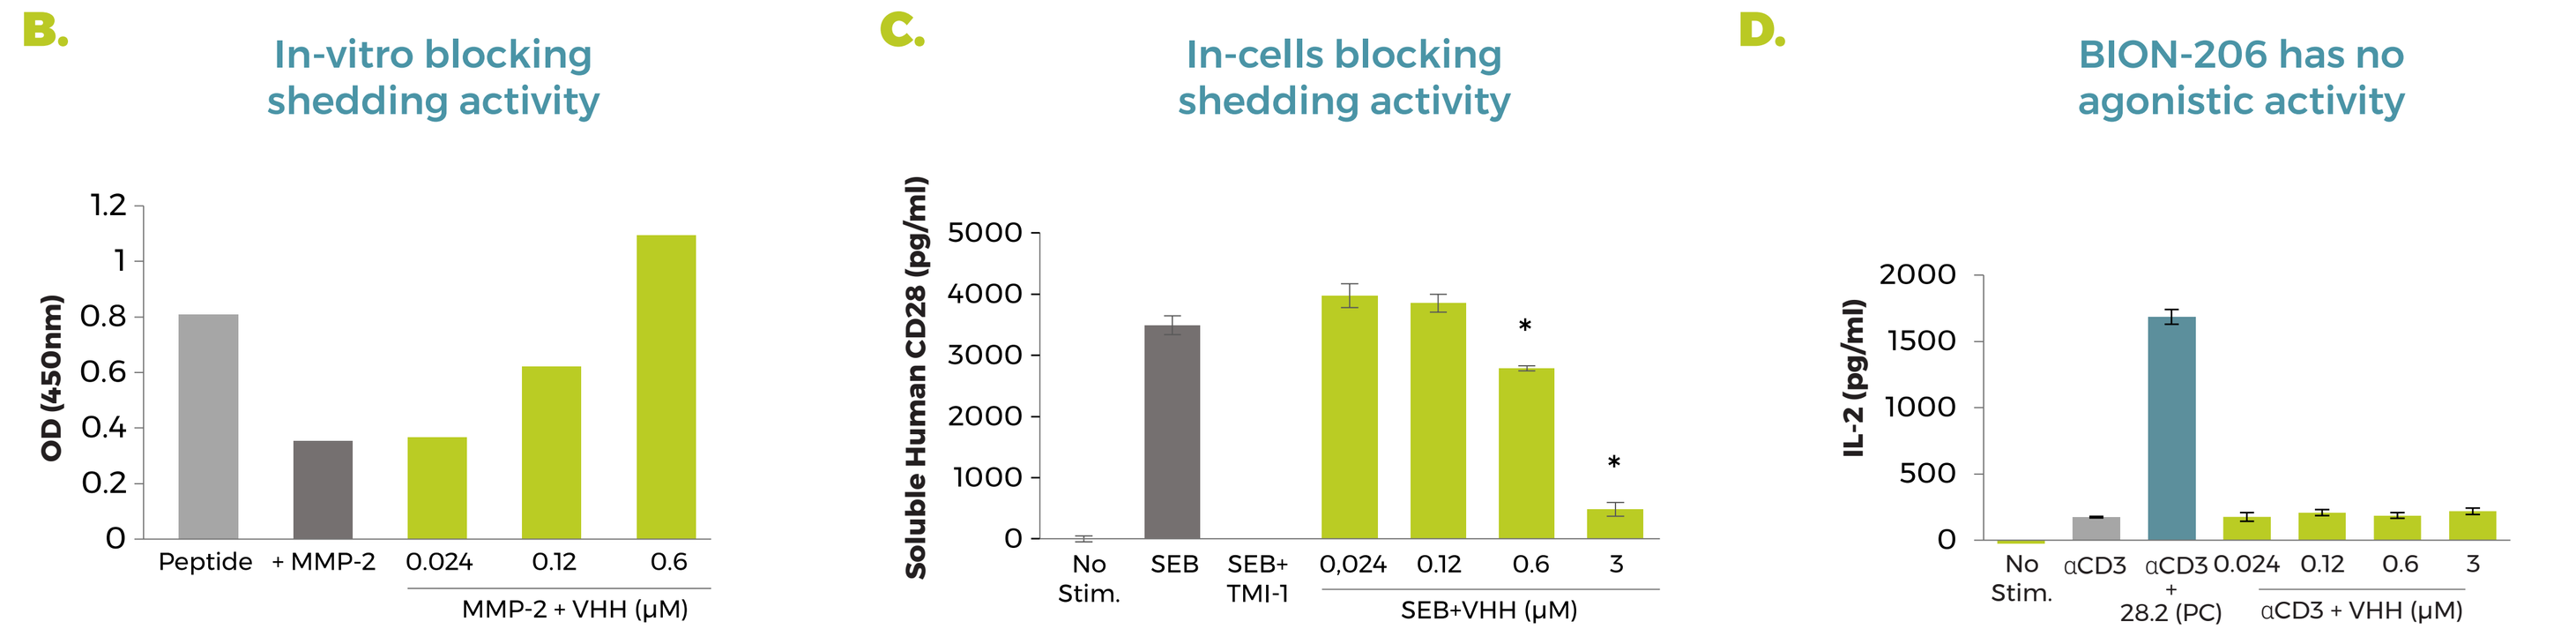
B.
In-vitro blocking shedding activity
OD (450nm)
Peptide
+ MMP-2
MMP-2 + VHH (µM)
C.
In-cells blocking shedding activity
Soluble Human CD28 (pg/ml)
No Stim.
SEB
SEB+ TMI-1
SEB+VHH (µM)
D.
BION-206 has no agonistic activity
IL-2 (pg/ml)
αCD3
αCD3 + 28.2 (PC)
αCD3 + VHH (µM)

VHH phage libraries constructed from a B cell repertoire of naïve and CD28 immunized Llamas were screened for anti CD28-shedding activity. A lead clone was characterized for binding to human CD28 stalk region by various methods (data not shown and A), in-vitro blocking of MMP-2 cleavage of human CD28 stalk region peptide (B), inhibition of CD28 shedding in T cells stimulation assays (C) and to not hold any agonistic effect on CD28 receptor (D). PC- positive control of anti-human CD28 clone 28.2. Soluble CD28 was detected using proprietary in-house ELISA. * P<0.05, unpaired student T.test compared to stimulation only.

10 Conclusions

- **We describe a novel mechanism in the CD28 pathway involving the proteolytic shedding of CD28**
- **CD28 shedding occurs upon T cell activation and causes the release of a dimeric soluble CD28 decoy receptor**
- **Soluble CD28 inhibits T cell activity and counteracts anti-PD1 activity**
- **High levels of sCD28 were found in cancer patients' blood, which were associated with response to PD-1 blockade**
- **A lead VHH antibody has been characterized and shown to bind specifically to CD28's proteolytic site, blocking CD28 cleavage with no antagonistic or agonistic activity**